Development of an RNA-based Diagnostic Platform Based on the Tumor Microenvironment Dominant Biology

Kristen Strand-Tibbitts[1], Kerry Culm-Merdek[1], Luka Ausec[2], Matjaz Zganec[2], Miha Stajdohar[2], Jeeyun Lee[3], Laura Benjamin[1] and Rafael Rosengarten[2]

[1] OncXerna Therapeutics, Inc. 300 Fifth Ave, Suite 6040, Waltham, MA 02451
[2] Genialis, Inc. 177 Huntington Ave, Suite 1703, Boston, MA 02115
[3] Samsung Medical Center, Sungkyunkwan University School of Medicine, Irwondong 50 Kangnamgu, Seoul, Korea 135-710

1 Background

Investigations of cancer biology have revealed a complex web of interactions between tumor cells and the surrounding non-tumor stroma that constitute the tumor microenvironment (TME). The TME regulates tumor growth by providing a physical substrate, nutrients and oxygen, and can regulate the surveillance and potency of the host immune system. Anti-cancer therapies have been developed to target drivers of tumor progression within the TME with differing degrees of success. These include therapies that disrupt angiogenesis or boost immune cell activity to promote tumor cell killing.

OncXerna is taking an approach of matching patients with TME-targeting therapies by assessing the dominant biological process in each patient, such that a patient with strong tumor angiogenesis will be matched to an anti-angiogenic drug while a patient with an immune response that is struggling can be matched to a therapy designed to enhance immune activity (**Figure 1**). NGS methods such as RNA-seq can be effective at describing patterns of biological activity within the tumor and its surrounding tissue.

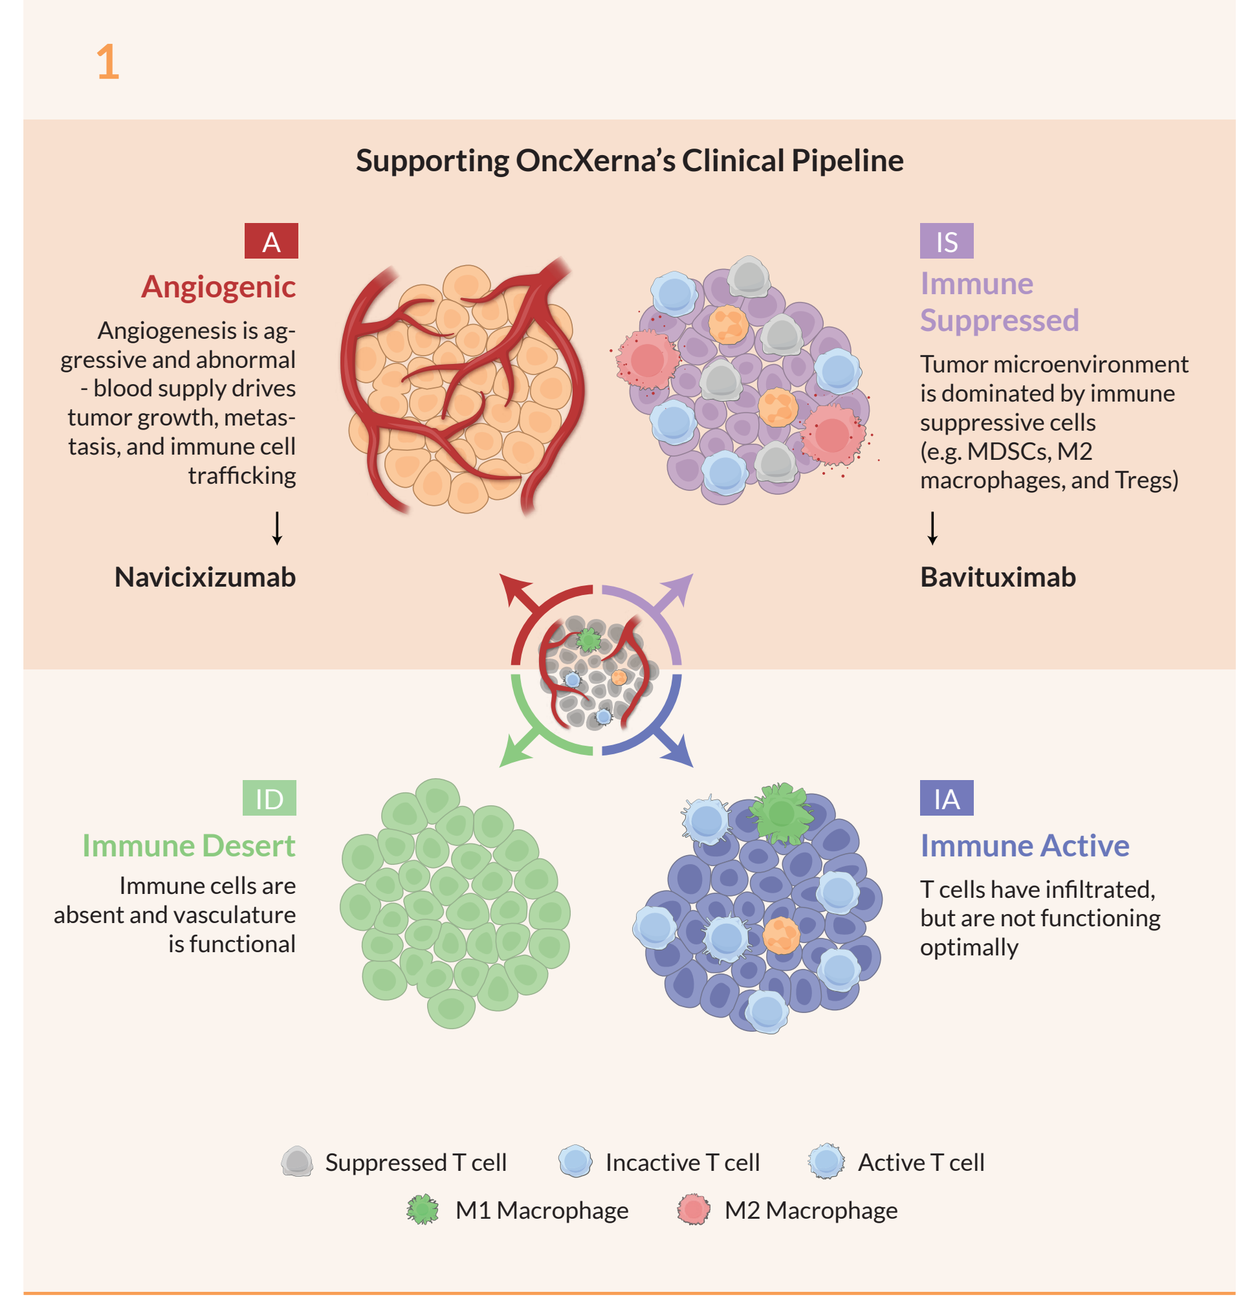
1
Supporting OncXerna's Clinical Pipeline
A
Angiogenic
Angiogenesis is aggressive and abnormal - blood supply drives tumor growth, metastasis, and immune cell trafficking
Navicixizumab
IS
Immune Suppressed
Tumor microenvironment is dominated by immune suppressive cells (e.g. MDSCs, M2 macrophages, and Tregs)
Bavituximab
ID
Immune Desert
Immune cells are absent and vasculature is functional
IA
Immune Active
T cells have infiltrated, but are not functioning optimally
Suppressed T cell
Incactive T cell
Active T cell
M1 Macrophage
M2 Macrophage

Figure 1. TME Panel-1 Identifies Four Dominant Biology Subgroups
Gene signatures representing dominant biologies of the stroma defined four tumor microenvironment (TME) phenotypes: Angiogenic (A), Immune Suppressed (IS), Immune Active (IA) and Immune Desert (ID). Previous work reported at SITC 2019 showed these phenotypes are independent of disease stage or demographics, and that the TME phenotypes confer distinct prognostic risk.

In order to identify patients whose TME biology is most susceptible to particular therapies, we have undertaken development of a machine-learning based diagnostic, herein TME Panel-1. We report results from the construction, training and validation of the TME Panel gene set and algorithm. The model was tested on data from gastric cancer patients treated with approved drug regimens and performed as well as existing industry-standards biomarkers. We retrospectively applied the model to data from two ongoing clinical trials of OncXerna's investigational drugs bavituximab and navicixizumab, demonstrating the ability to enrich for responses and identify non-responses. These results contribute towards a diagnostic platform that could possibly **1)** be tailored to specific disease sites and drugs; **2)** accommodate the workflow of clinical sites and testing labs; and **3)** provide actionable results to facilitate decision-making by clinicians, caregivers, patients and their families.

2 Methods

The gene signatures identified previously (Strand-Tibbitts, 2019. SITC: Working Towards Precision Medicine of the Tumor Microenvironment) were used as a starting point in development of a first generation diagnostic algorithm, TME Panel-1. All microarray and RNA-sequencing data were either reprocessed or analyzed anew using Genialis Expressions software. Gene expression values were normalized to transcripts per million and quantile transformed to a near normal distribution. Each gene in the signatures was evaluated as a potential model feature based on the consistency of its distribution of expression across different platforms (microarray, RNA-seq, Edge-seq), and different public gastric cancer datasets (ACRG, TCGA, Singapore cohort). Genes that displayed quantitatively consistent expression distributions were retained for model training.

Various machine learning algorithms were trained on microarray data from ~300 gastric cancer patient samples (ACRG, see Box: **Gene Expression Datasets Used in Study**). Performance was evaluated by the ability to predict clinical responses in an independent RNA-seq dataset (Samsung CPI, see Box below). The highest performing model was an artificial neural network (ANN) consisting of an input layer (feature gene set), hidden layer (network nodes that learned the weights of the feature genes), and output layer (four TME phenotype classes described in **Figure 1**). Optimization steps included further feature reduction, regularization of the model hyperparameters and exploration of biomarker thresholds based on various clinical endpoint and response metadata.

The Samsung CPI dataset was reanalyzed with the optimized ANN model. Subjects were called biomarker positive or negative based on probability of their TME phenotype call—e.g. Immune Active (IA) subjects could be biomarker positive for a checkpoint inhibitor. Biomarker predictions were evaluated by comparing the objective response rate (ORR)—examined across a battery of clinical attributes—of all-comers versus the TME Panel classifications. The TME Panel biomarker was compared head to head with other industry standard biomarkers, such as PD-L1 CPS>1 or MSI-High, based on metrics for accuracy, AUC ROC, sensitivity, specificity, positive predictive value and negative predictive value.

To assess the potential to employ TME Panel-1 as a diagnostic across tumor sites, 1100 samples from gastric, ovarian and colorectal cancer were analyzed by exome RNA-seq, and TME phenotypes assigned by the ANN model. To further test the applicability of the model across drug regimen and cancer types, the gastric cancer derived ANN model was applied retrospectively to RNA-seq data from OncXerna's ongoing clinical trials—bavituximab + pembrolizumab in gastric cancer, and navicixizumab + paclitaxel in ovarian cancer.

Gene Expression Datasets Used In Study

The following datasets were used in the development, training, testing, validation and clinical application of the TME Panel-1 biomarker

ACRG (Asian Cancer Research Group)
- Gastric cancer subjects (N=300) were second line or beyond, receiving prior chemotherapy and/or radiation
- Affymetrix microarray; GEO GSE62254, GSE62717; Cristescu et al 2015

TCGA (The Cancer Genome Atlas)
- Gastric cancer subjects (N=388) were a mixture of multiple lines of treatment
- RNA-seq; Data at portal.gdc.cancer.gov; Cancer Genome Atlas Research Network 2014

Singapore
- Gastric cancer subjects (N=192) were a mixture of multiple lines of treatment
- Affymetrix microarray platforms; GEI (GSE15459); Lei et al 2013

Samsung CPI
- Subjects with gastric and GEJ cancer, mixed prior treatment history, 100% Asian demographic
- Treated with anti-PD-L1 checkpoint inhibitors pembrolizumab (56) or nivolumab (17)
- RNA-seq (N=73); Data are unpublished

ONCG100 / Bavi
- Ongoing single arm phase II trial (NCT04099641)
- Subjects with gastric (2/3) and GEJ (1/3) cancer, predominantly 2nd and 3rd line, 45% Asian + 55% non-Asian
- Treated with a combination of pembrolizumab and bavituximab
- RNA-seq (N=38 to date); Data are unpublished

B83-002 / Navi
- Single arm phase 1b study of 4+ line platinum resistant patients with ovarian cancer treated with the combination of navicixizumab/paclitaxel
- RNA-seq (subset N=30); Data are unpublished

3 Validating TME Panel-1 with CPI Treatment Data

To validate that TME Panel-1 assigns patients to therapeutically relevant phenotype classes, the ANN model was used to classify subjects from the Samsung CPI dataset. The biomarker hypothesis was that Immune Active (IA) subjects are most likely to show response to CPI treatment.

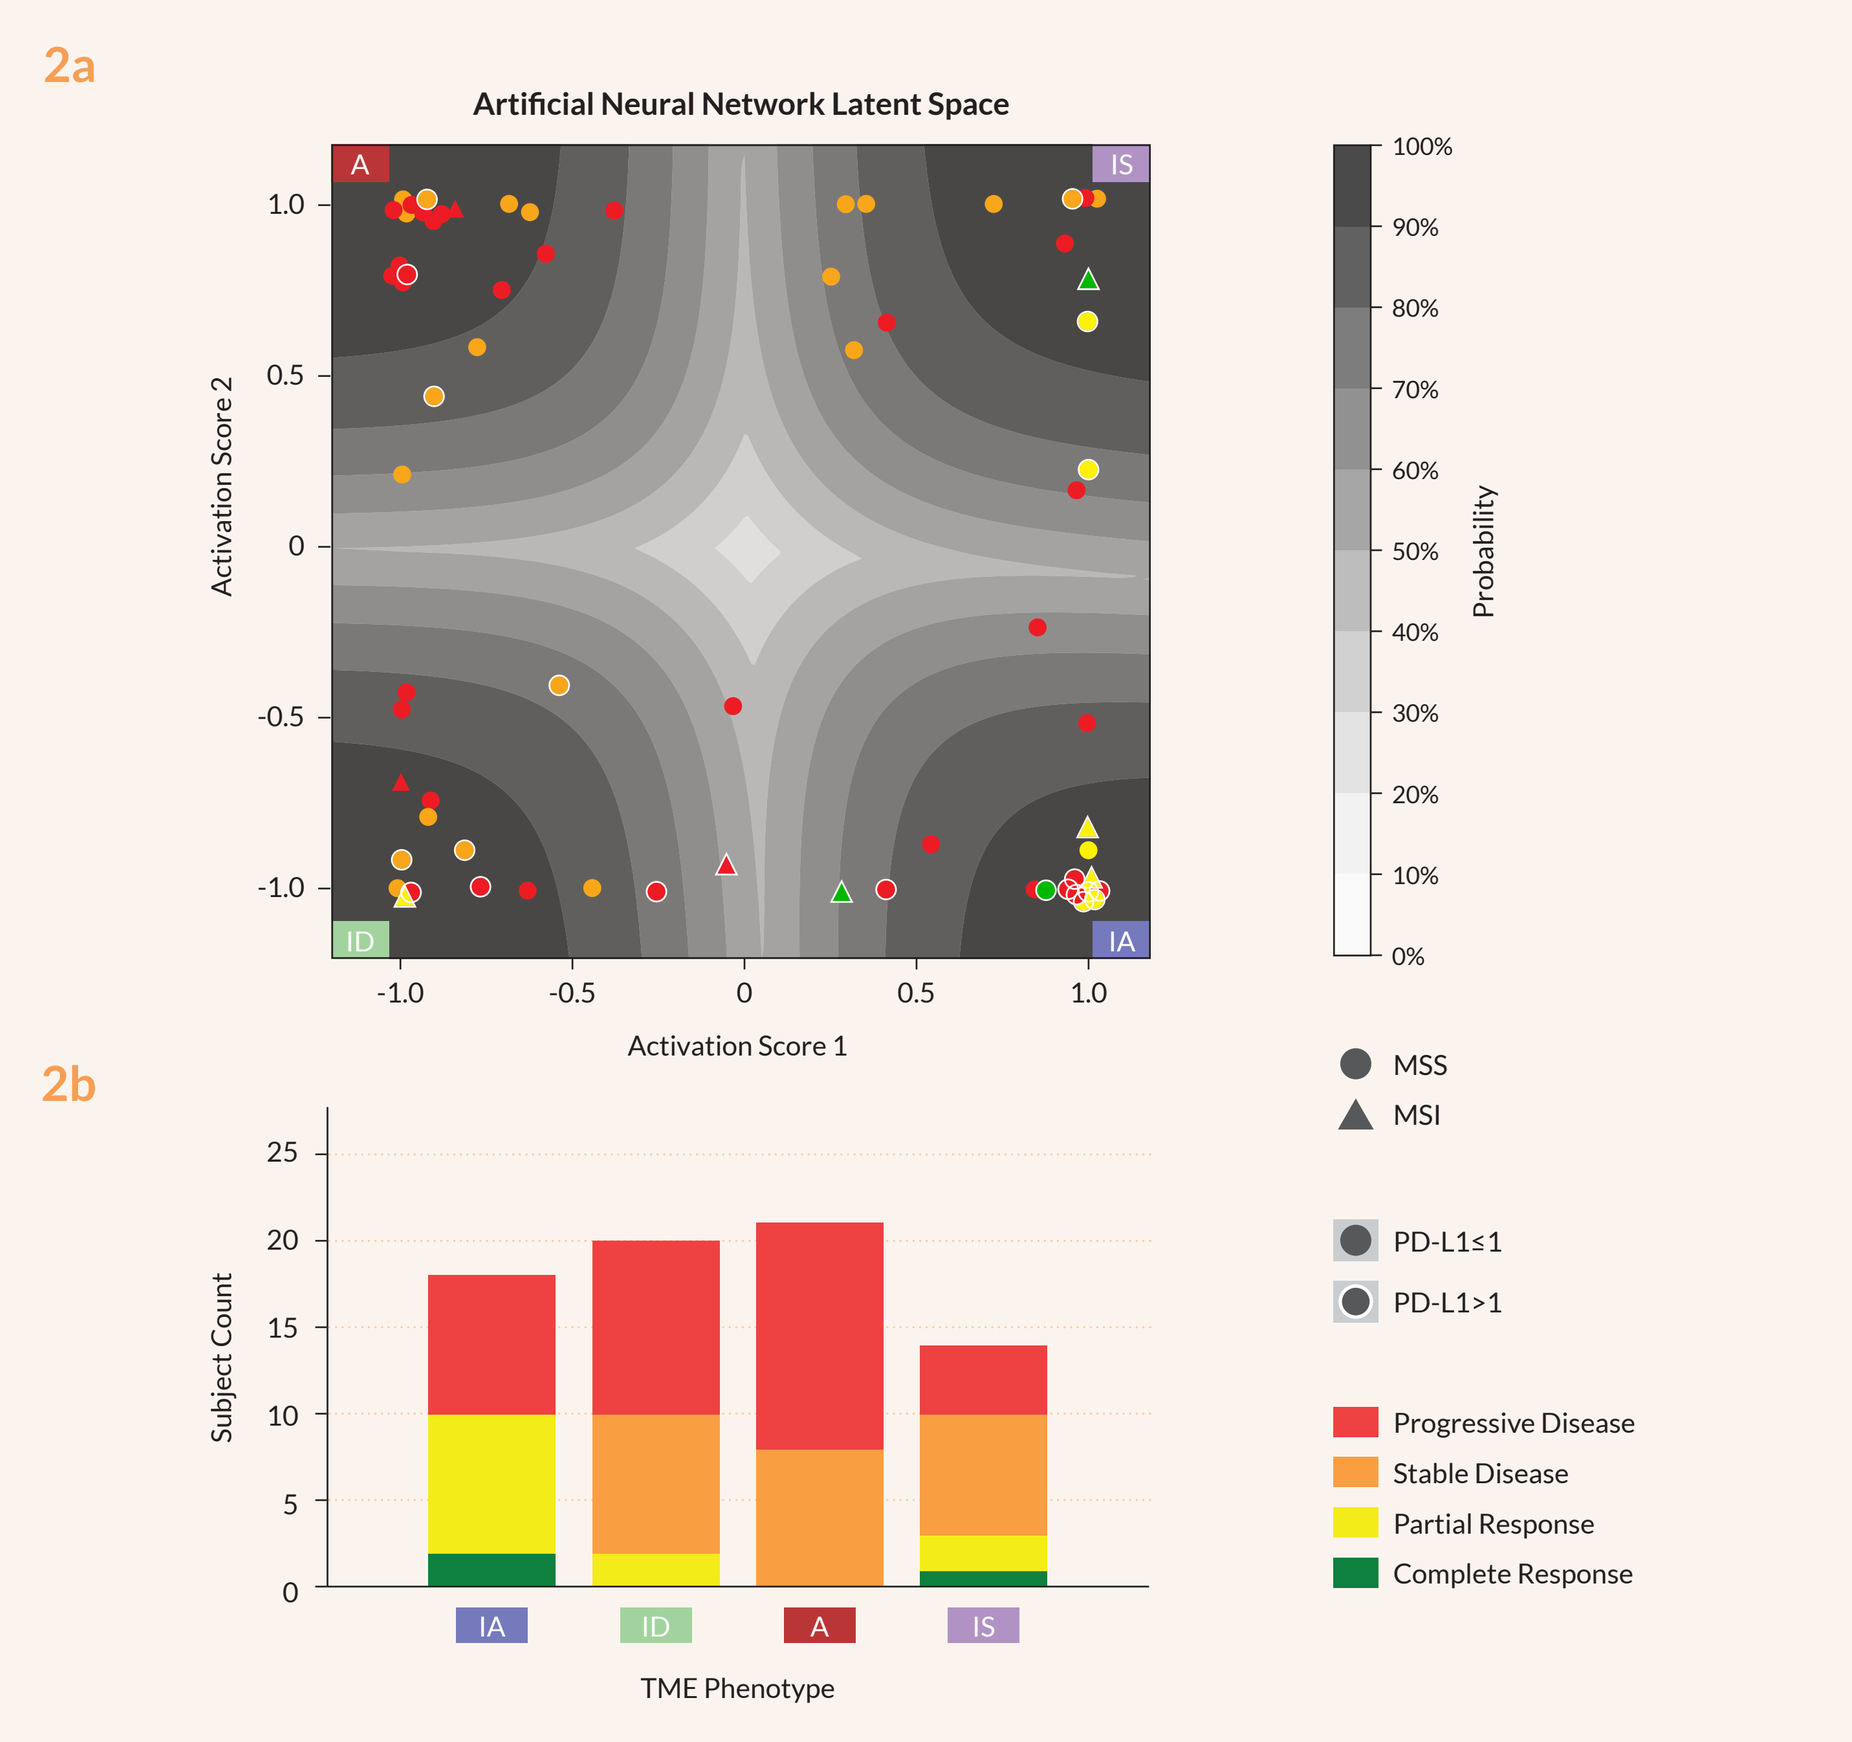
2a
Artificial Neural Network Latent Space
A
IS
ID
IA
Activation Score 1
Activation Score 2
Probability
100%
90%
80%
70%
60%
50%
40%
30%
20%
10%
0%
MSS
MSI
PD-L1≤1
PD-L1>1
2b
Subject Count
TME Phenotype
IA
ID
A
IS
Progressive Disease
Stable Disease
Partial Response
Complete Response

Figure 2. Response to CPI Treatment Predominant in Immune Active TME Class of Gastric Cancer Subjects
Figure 2a. *Biomarker positive thresholds were evaluated at increasingly stringent probability levels, though for this dataset, the class assignments themselves performed notably well (**Table 1**). Clinical metadata were marked as colors or glyphs, as noted in the legend, to facilitate the analysis of the relationship between TME phenotype and various readouts, e.g. best objective response, PD-L1 CPS, MSI status, etc., (summarized in **Table 1**).*
Figure 2b. *The distribution of tumor responses to CPIs as a function of TME phenotype. Best objective response is denoted based on RECIST criteria as complete response, partial response, stable disease or progressive disease. Given the anti-PD-L1 mechanism of action, and the predictive basis of the algorithm, , the largest number of responses was observed in the Immune Active (IA) TME phenotype class, whereas the Angiogenic (A) class saw no responses. The IA class, meanwhile, included no stable disease subjects, while the IS group was mostly stable disease in addition to its three responses.*

The flexibility of the approach to define biomarker groups from the ANN phenotype classifications lent itself to investigating the relationships between tumor response rate and other clinical variables in the various TME classes. The unselected cohort recorded a modest ORR of 18%, while the IA class yielded an ORR of 44%, though it missed five responses overall.

Standard biomarkers like PD-L1 > 1 identified nearly all of the true responses, but also included many non-responses (12/40 of all subjects). Meanwhile, no responses were observed among the 29 subjects with PD-L1 < 1. The goal is not necessarily to propose a better biomarker for checkpoint inhibitors, rather to demonstrate that the TME Panel captures biologically and therapeutically relevant characteristics of the patient population, perhaps missed by the standard molecular markers. Exploring response rates in other molecular categories, e.g. MSS, may help direct future treatment to underserved patient subsets.

Table 1. Retrospective Analysis of Best Objective Response from Gastric Cancer Patients Treated with a CPI
*Objective response rates (ORR) to CPI treatment, grouped by the ANN-classified TME phenotypes, were studied in relation to other clinical characteristics. Neutrophil-to-Lymphocyte Ratio (NLR) was divided at the median (</>4) observed for the ONCG100 study (**Figure 4**). PD-L1 was evaluated at composite positive score thresholds of 1 and 10, as determined by IHC.*

ORR		N, %		NLR ≤ 4		PD-L1 <1		PD-L1 >1		PDL1 <10		PD-L1 > 10		MSI		MSS	
All subjects	N=73	13/73	18%	11/56	20%	0/29	0%	12/40	30%	4/52	8%	8/17	47%	5/8	63%	8/65	12%
IA	N=18	8/18	44%	6/14	43%	0/3	0%	8/14	57%	3/10	30%	5/7	71%	3/3	100%	5/15	33%
IS	N=14	3/14	21%	3/13	23%	0/8	0%	3/6	50%	1/12	8%	2/2	100%	1/1	100%	2/13	15%
ID	N=20	2/20	10%	2/14	14%	0/5	0%	1/12	8%	0/12	0%	1/5	20%	1/3	33%	1/17	6%
A	N=21	0/21	0%	0/15	0%	0/13	0%	0/8	0%	0/18	0%	0/3	0%	0/1	0%	0/20	0%

4 Applying TME Panel-1 to Bavituximab

OncXerna is developing bavituximab—a novel, first-in-class phosphatidylserine (PS) inhibitor that reverses T-cell exhaustion and restores tumoricidal macrophage function. Combined use of checkpoint inhibitors and antibodies that block PS signaling to immune cell receptors may produce greater therapeutic benefit than either monotherapy alone.

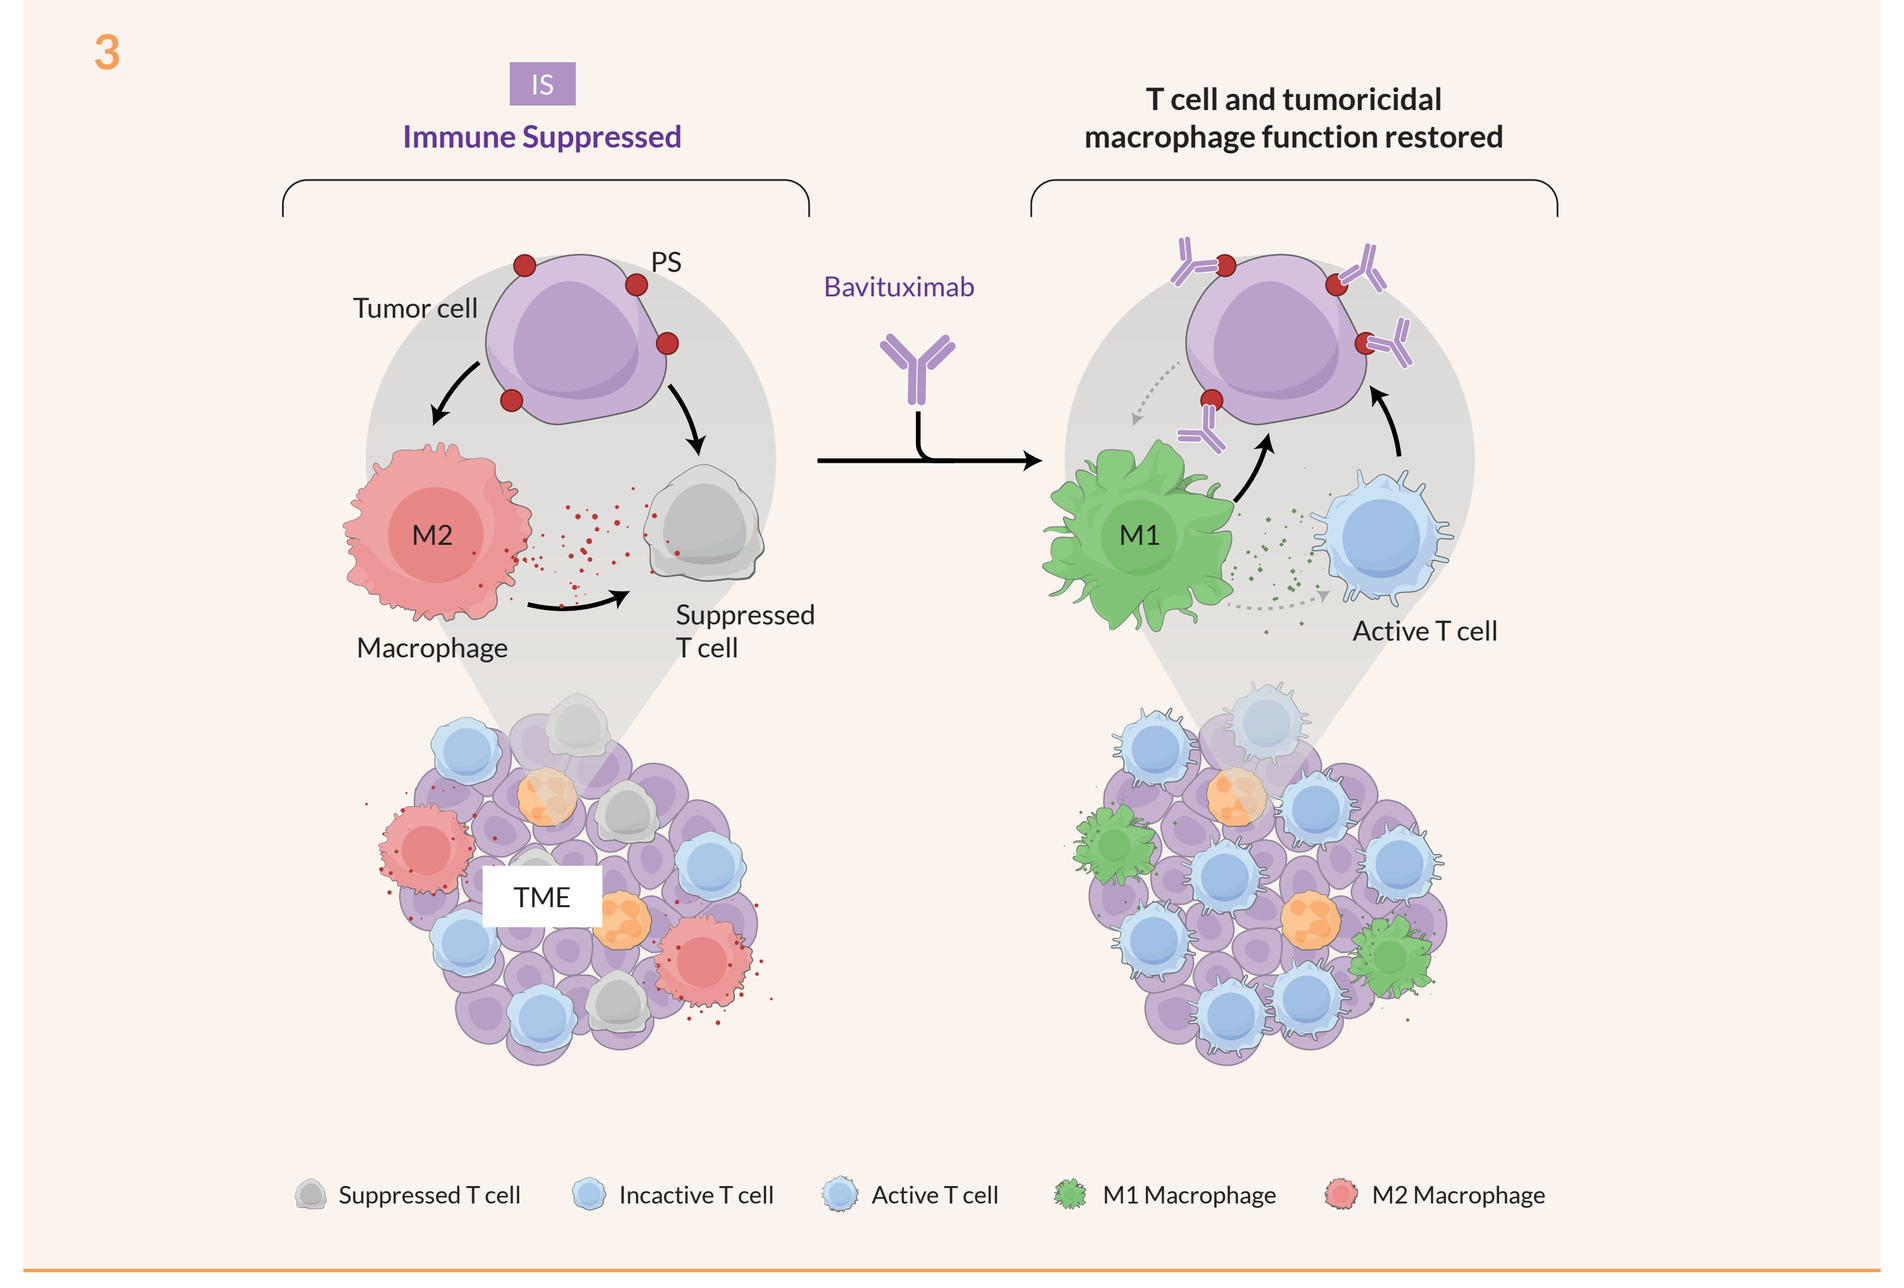
3
IS
Immune Suppressed
T cell and tumoricidal macrophage function restored
Tumor cell
PS
Bavituximab
M2
Macrophage
Suppressed T cell
M1
Active T cell
TME
Suppressed T cell
Incactive T cell
Active T cell
M1 Macrophage
M2 Macrophage

Figure 3. Bavituximab Reverses Immune Suppression
PS is a phospholipid located on the inner surface of the cell membrane. In tumor cells, PS relocates to the outer surface where it acts as an immunosuppressive ligand for multiple immune receptors, including TIM/TAM (T cell immunoglobulin and mucin domain 3) receptors. Bavituximab reverses immune suppression by inhibiting PS (TIM/TAM) signaling, activating immune cells. The biomarker hypothesis based on this MOA is that patients who are IS could show enhanced responses to CPIs when treated with bavituximab.

Since the goal of creating and optimizing the TME Panel is to guide future clinical development, data was interrogated from an interim look at the ongoing ONCG100 trial in order to examine the relationship of biomarker class to patient health, treatment duration and other clinical endpoints. ONCG100 is a phase 2 open label study of bavituximab and pembrolizumab in advanced gastric and GEJ cancer patients, for whom tumor biopsies were analyzed by IHC for PD-L1 and RNAseq for analysis of the TME Panel. Patients in both the IA or the IS biomarker subgroup would be expected to benefit from this combination. ONCG100 Group 1 includes patients that are CPI naïve and Group 2 includes patients with durable responses or stable disease on prior CPI who relapsed before joining this study.

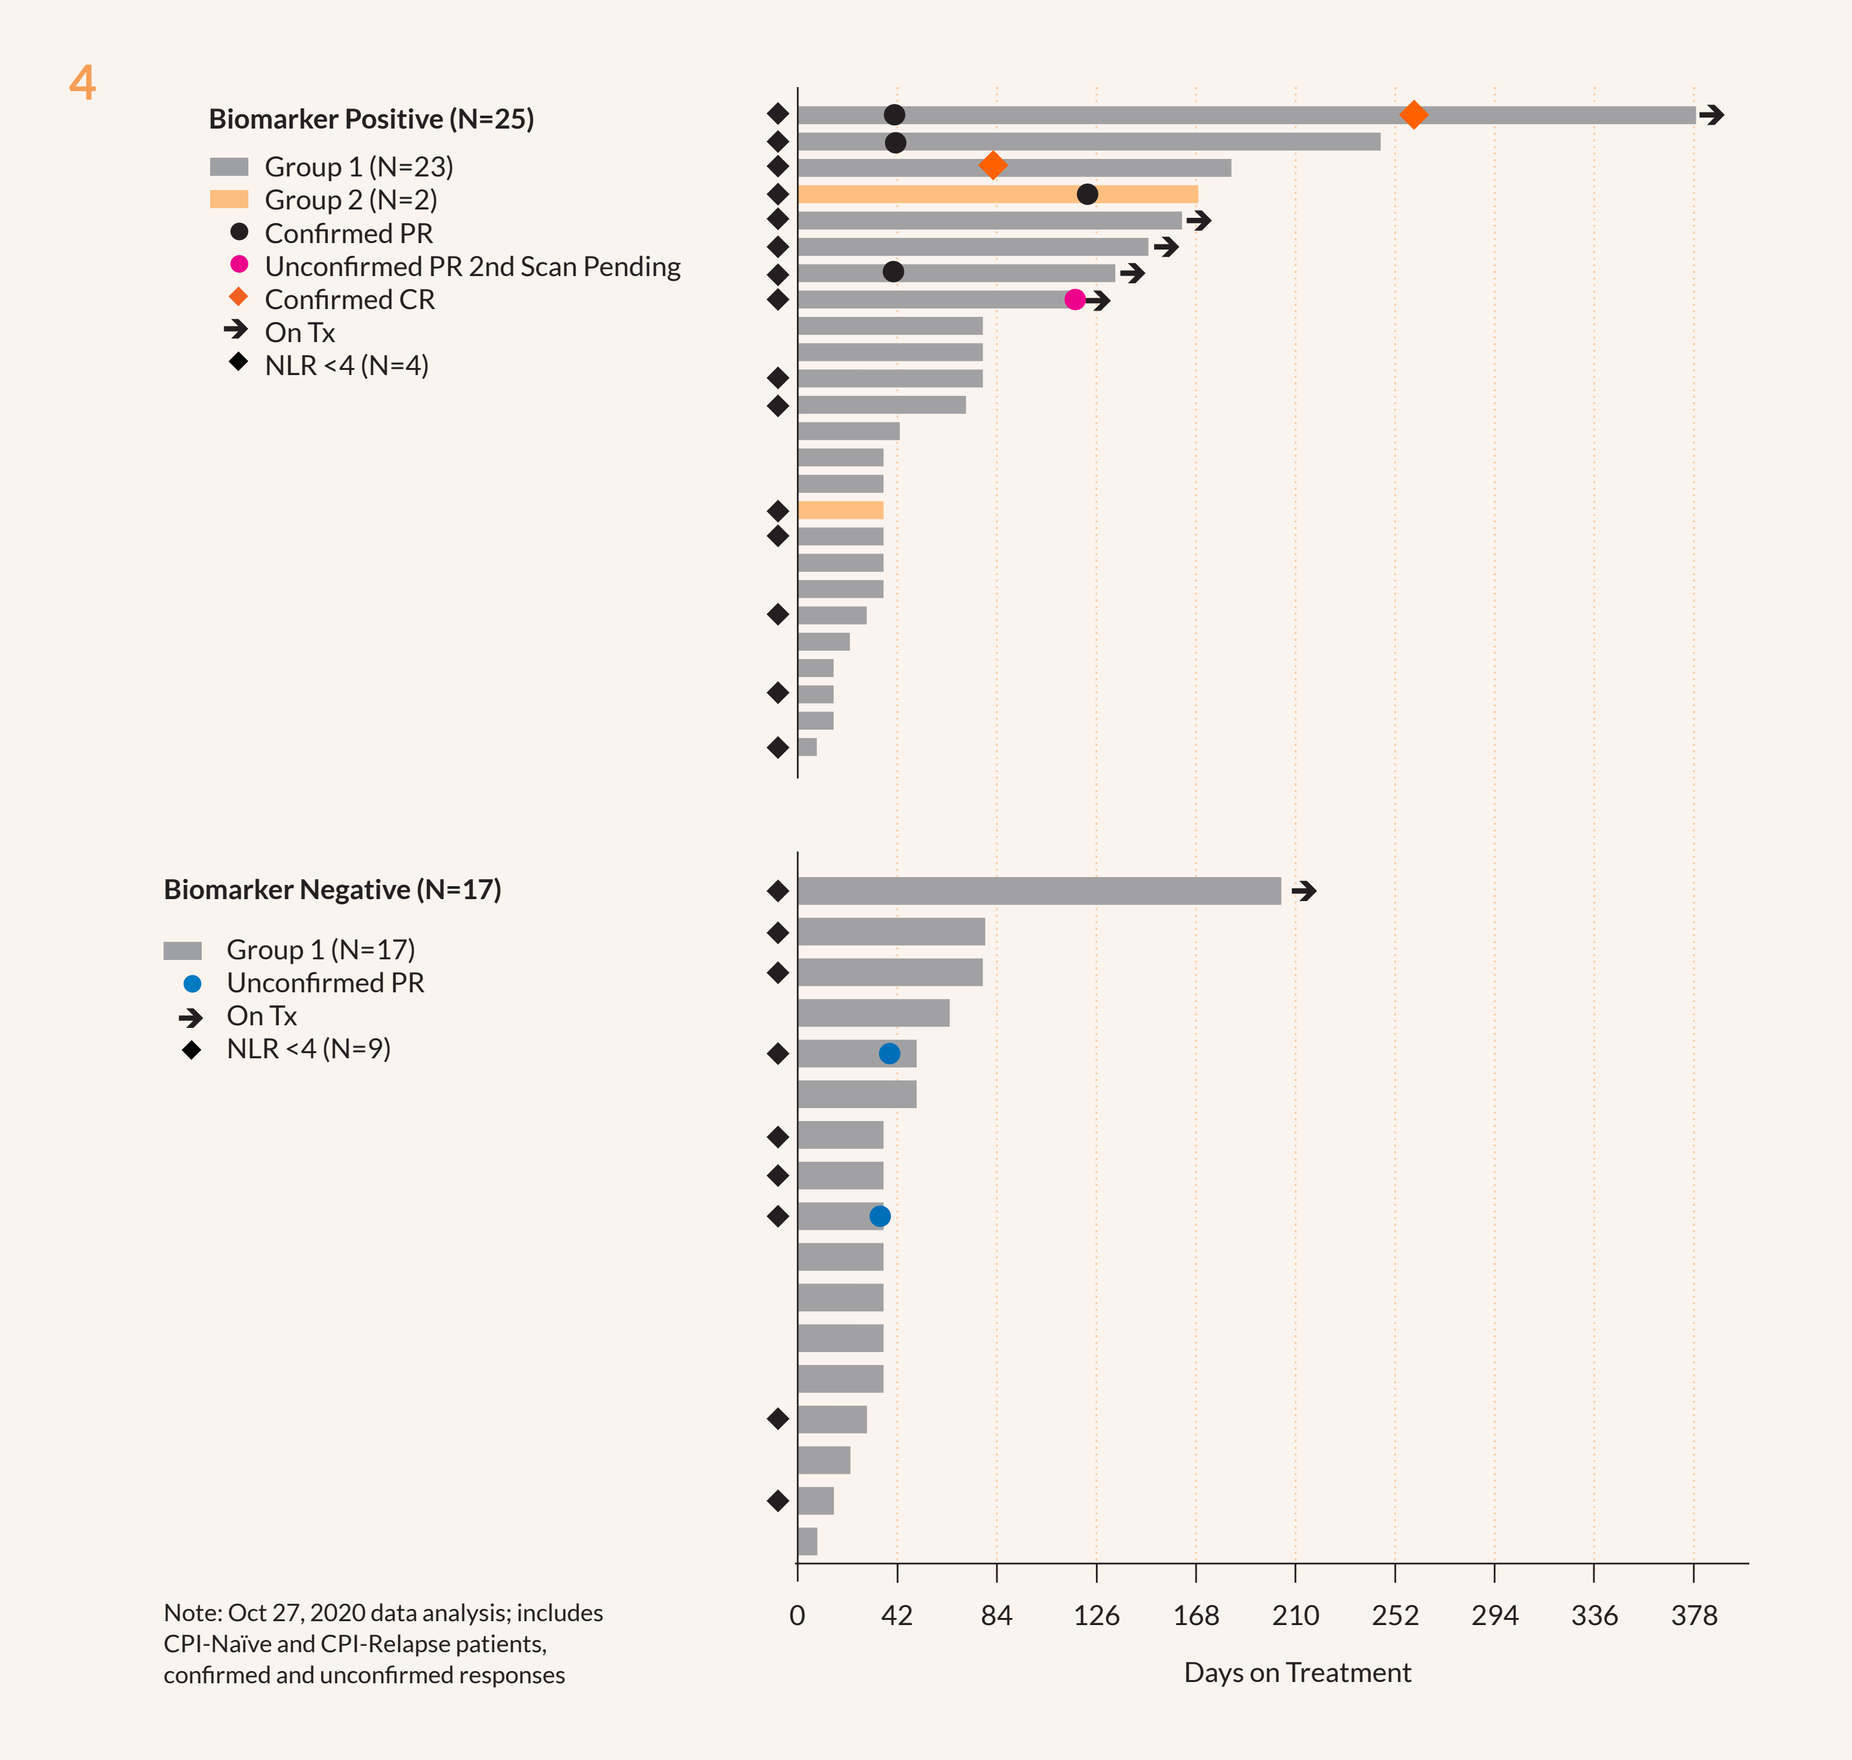
4
Biomarker Positive (N=25)
Group 1 (N=23)
Group 2 (N=2)
Confirmed PR
Unconfirmed PR 2nd Scan Pending
Confirmed CR
On Tx
NLR <4 (N=4)
Biomarker Negative (N=17)
Group 1 (N=17)
Unconfirmed PR
On Tx
NLR <4 (N=9)
Note: Oct 27, 2020 data analysis; includes CPI-Naïve and CPI-Relapse patients, confirmed and unconfirmed responses
0
42
84
126
168
210
252
294
336
378
Days on Treatment

Figure 4. Biomarker Positive Class Captures Patients with Improved Responses and Durability
Tumor response is scored based on RECIST criteria. More responses were observed thus far in the biomarker positive group, with 2 confirmed complete responses (CR), 3 confirmed partial responses (PR), and 1 unconfirmed partial response (awaiting second scan results). In the biomarker negative group, the two unconfirmed responses progressed on their next scan. Baseline CBC values for neutrophil-to-lymphocyte ratio (NLR) were divided at the median (</> 4).

Table 2. Difference in Tumor Response Between Biomarker Positive vs. Negative Patients in ONCG100 Trial
A clear difference was observed between biomarker positive versus negative, with all confirmed tumor responses to bavituximab plus pembrolizumab occurring in the biomarker positive group (IA+IS). Objective response rate (ORR) was tabulated as all complete response (CR) + partial response (PR) / total subjects. Disease control rate (DCR) included CR + PR + stable disease (SD). Progressive disease (PD) constituted a greater fraction of subjects in the biomarker negative group. These results support the application of the TME Panel to derive a diagnostic algorithm for prospective trials.

Tumor Response[1]	Biomarker Positive (N=22)[1]	Biomarker Negative (N=16)[1]
ORR[2]	27%	0%
DCR	45%	13%
CR[2]	9%	0%
PR[2]	18%	0%
SD	18%	13%
PD	55%	88%

1 CPI-Naïve and CPI-Relapse.
2 Confirmed responses + one unconfirmed response that had not yet had 2nd Scan at the time of this analysis. Patients with a second scan that failed to confirm response were not included.

Patients with confirmed response to treatment with bavituximab and pembrolizumab were observed entirely among biomarker positive (IA + IS) subjects. Next, biomarker performance on the combination treatment dataset next was compared to its performance on the Samsung CPI monotherapy dataset on which the model was initially validated.

Table 3. Comparing Biomarker Performance on ONCG100 versus Samsung CPI Cohort
The ANN model was applied to RNA-seq data from the ONCG100 clinical trial, and performance of the biomarker assessed retrospectively. Best objective response was used to score the model, with CR and PR considered clinical benefit. The model performance on the ONCG100 data was compared to its performance on the Samsung CPI cohort, and to the predictive performance of industry-standard diagnostic analytes such as PD-L1 CPS (combined positive score) and MSI-H (microsatellite instability-high). The ONGC100 biomarker threshold was set based on the bavituximab intent to treat population, that is, patients with NLR<4 and classified as IA or IS. For the Samsung dataset, all subjects in the IA phenotype class were considered biomarker positive. The TME biomarker performed as well as standard CPI biomarkers, and demonstrated similarly high utility in the ONCG100 combination therapy cohort.

	Biomarker Positive	ACC	ROC AUC	Sensitivity	Specificity	PPV	NPV
ONCG100	TME Panel IA + IS	0.58 (22/38)	0.75	1.00 (6/6)	0.50 (16/32)	0.27 (6/22)	1.00 (16/16)
	TME Panel IA + IS NLR < 4	0.64 (14/22)	0.75	1.00 (6/6)	0.50 (8/16)	0.43 (6/14)	1.00 (8/8)
Samsung CPI Cohort	TME Panel IA	0.79 (58/73)	0.72	0.6 (8/13)	0.8 (50/60)	0.44 (8/18)	0.91 (50/55)
	PD-L1 CPS>1	0.75 (55/73)	0.79	0.85 (11/13)	0.73 (44/60)	0.41 (11/27)	0.96 (44/46)
	MSI-H	0.85 (62/73)	0.67	0.38 (5/13)	0.95 (57/60)	0.62 (5/8)	0.88 (57/65)

Accuracy (ACC): Number of correct predictions / Total number of predictions
Receiver Operating Characteristics Area Under the Curve (ROC AUC): Degree to which model is capable of distinguishing between classes
Sensitivity: True biomarker responses / Total actual responses
Specificity: True biomarker non-responses / Total actual non-responses
Positive Predictive Value (PPV): True biomarker responses / Total predicted biomarker responses
Negative Predictive Value (NPV): True biomarker non-responses/ Total predicted biomarker non-responses

To more directly assess the hypothesis that bavituximab sensitizes patients to check-point inhibition, and to identify population subsets that stand to benefit the most, response rates were compared between the ONCG100 combination and pembrolizumab monotherapy from the KEYNOTE-059 trial. KEYNOTE-059 was a global study with 47% of subjects in the US, and 29% either in East Asia or of that diaspora. Subjects were split evenly between gastric and GEJ subjects, 51% in the 3rd line of therapy. In contrast to the model validation exercise using the demographically homogenous Samsung CPI cohort, the KEYNOTE study more closely resembles that of the ONCG100 trial.

Table 4. Strong Activity Observed in Patients with Low Historical Responses to Keytruda
In the KEYNOTE-059 trial, response rates were tabulated based on a number of biomarkers and molecular characteristics. Two subgroups, MSS and PD-L1 CPS<1, underperformed the all comers ORR. In the ONCG100 study, meanwhile, MSS and PD-L1 CPS<1 subjects responded at more than 2-3 times the rate as with Keytruda monotherapy. Further, biomarker selected patients from the intent to treat population experienced an ORR more than 2x the ONCG100 all comers ORR, and more than 3x the KEYNOTE-059 all comers ORR.

	Keytruda single-arm (KEYNOTE-059[1])				Bavituximab + Keytruda (ONCG100) all comers[2,3]						
Clinical Benefit	Overall	MSS	PD-L1 CPS≥1	PD-L1 CPS<1	Overall (N=44)[4]	MSS (N=28)[4]	PD-L1 CPS≥1 (N=22)[4,5]	PD-L1 CPS<1 (N=10)[4,5]	NLR<4 (N=28)[4]	TME Panel (N=22)[4,6]	NLR<4 + TME Panel (N=14)[4,6]
ORR	12%	9%	16%	6%	20%	21%	18%	20%	32%	27%	43%
CR	2%	2%	3%	3%	5%	7%	0%	20%	7%	9%	14%

1 Phase 2 Keynote-059 Trial: Fuchs CS, JAMA Oncology May 2018 Volume 4, Number 5
2 One confirmed response still on treatment approaching one year on treatment
3 October 15, 2020 data analysis CPI-Naïve & CPI-Relapse
4 Confirmed and unconfirmed responses
5 Local CPS scores utilized when central lab scores were not available. PD-L1 values were available on 32 subjects.
6 RNAseq was available on 38 subjects

5 TME Panel-1 Pan-Cancer Application to Navicixizumab

The interactions between tumor and stoma govern progression of cancers in many different tissue types. Gastric cancer is by no means unique in the dominant biology captured by OncXerna's TME Panel-1. To test whether the TME Panel can classify subjects with other cancer sites, nearly 1100 samples—evenly distributed across three different diseases—were analyzed by RNA-seq and those subjects classified with the ANN model.

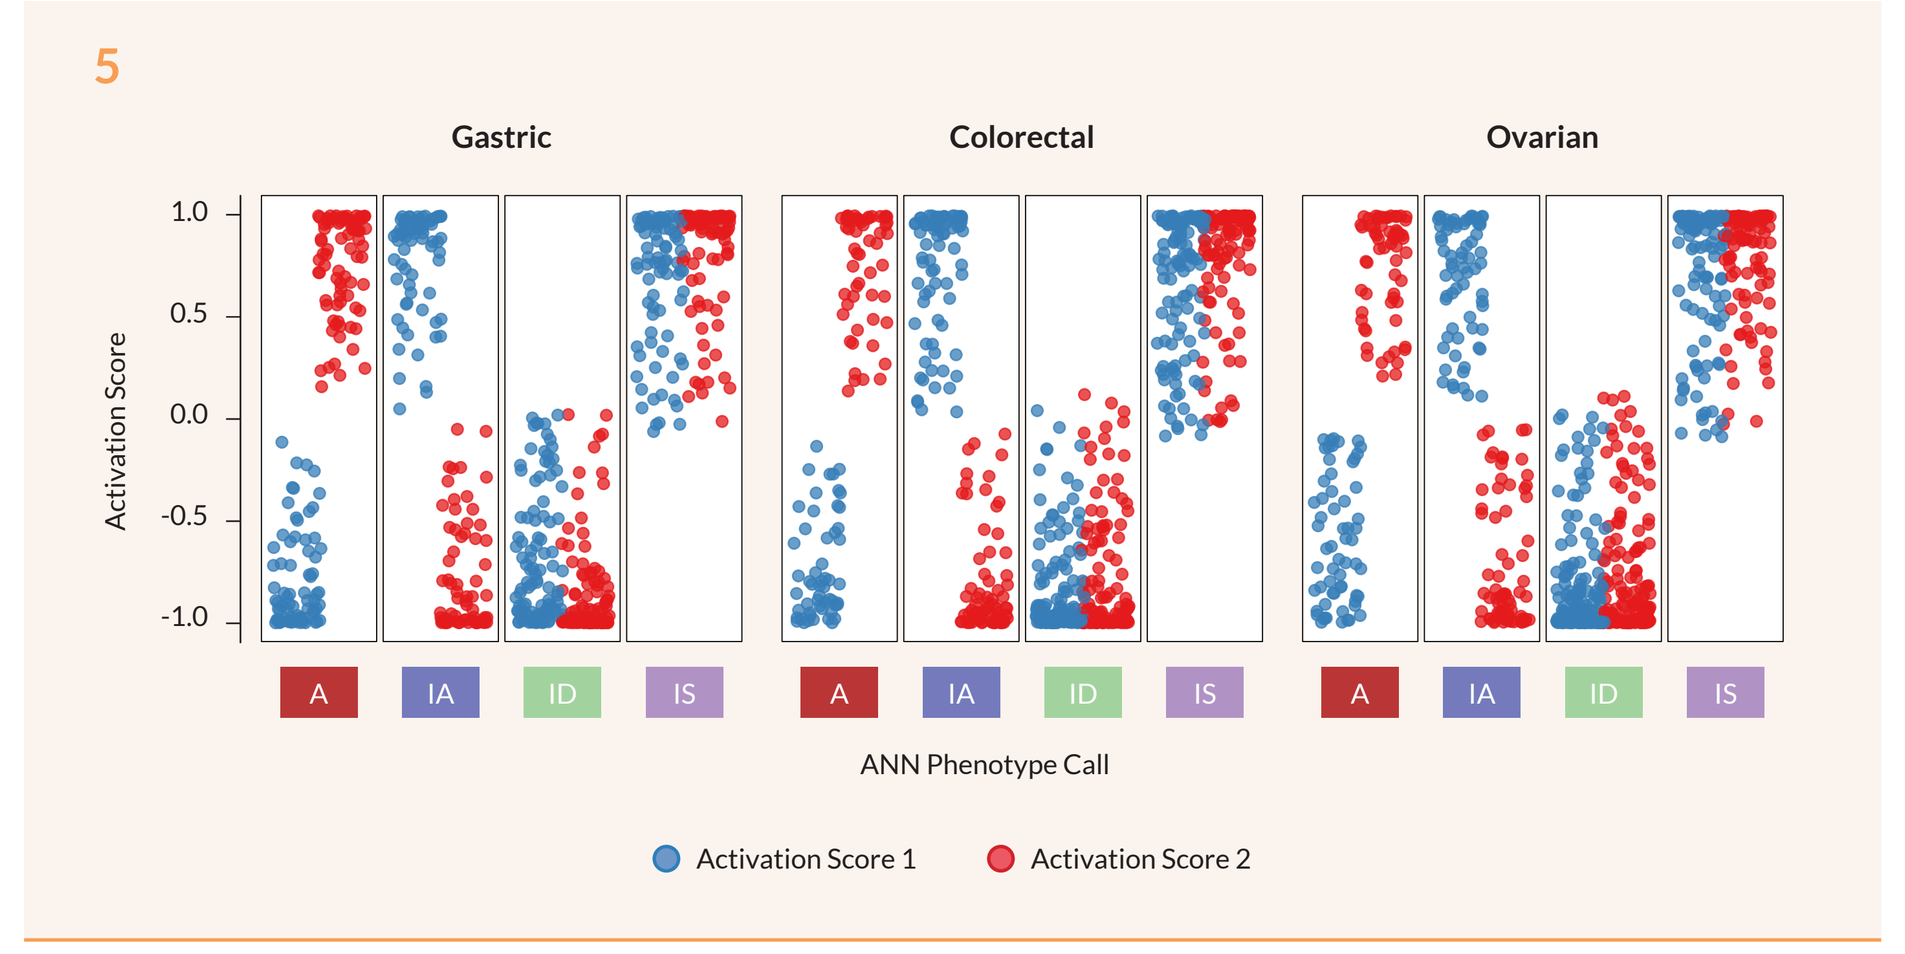
5
Gastric
Colorectal
Ovarian
Activation Score
1.0
0.5
0.0
-0.5
-1.0
A
IA
ID
IS
ANN Phenotype Call
Activation Score 1
Activation Score 2

Figure 5. TME Panel-1 Predicts Stromal Phenotypes Across Tumor Sites
Roughly 400 subjects from each of gastric, colorectal and ovarian cancer were assigned one of the four TME phenotypes by the ANN classifier. The activation scores of two characterized neurons—which generally weight immune (blue) and angiogenic (red) processes— are plotted on the vertical access for each subject. The corresponding phenotype predictions are annotated on the X-axis. The distribution of subjects across the four TME phenotypes was similar regardless of disease, suggesting the the classifier could be optimized, and TME Panel applied, for a variety of cancers.

In addition to bavituximab, OncXerna's pipeline includes an investigational drug with early evidence of efficacy in ovarian cancer—navicixizumab. Navicixizumab is a bispecific mAb targeting VEGF and DLL4, and thus impedes angiogenesis and stromal remodeling via the notch pathway. To test whether the TME Panel could identify gynecological cancer patients more likely to experience a response to therapy, RNA-seq data gathered during the phase 1b trial of navicixizumab + paclitaxel was analyzed retrospectively.

Table 5. TME Panel-1 Retrospectively Enriches for Responses to Navicixizumab
Among 23 subjects with available RNA-seq data and confirmed best objective response (RECIST), 70% of the biomarker positive group experienced an objective response, while only 31% of the biomarker negative group reported the same. Because the navicixumab mechanism of action is anti-angiogenic, subjects assigned to the A + IS phenotype class were considered biomarker positive. Objective response rate (ORR) was tabulated as all complete response (CR) + partial response (PR) / total subjects. Disease control rate (DCR) included CR + PR + stable disease (SD). Progressive disease (PD) constituted a greater fraction of subjects in the biomarker negative group, while no PD subjects were biomarker positive. The observed biomarker responses support applying the TME panel to further navicixizumab trials. Moreover, these results highlight the versatile nature of the TME panel, applicable to drugs of distinct MOA and tumors of distinct TME phenotype.

	Avastin-naïve and -experienced[1]	
Tumor Response	Biomarker Positive (N=10)	Biomarker Negative (N=13)
ORR	70%	31%
DCR	100%	69%
CR	0%	8%
PR	70%	23%
SD	30%	39%
PD	0%	31%

1 Of the 30 patients with tissue available for Biomarker analysis, 23 had confirmed responses.

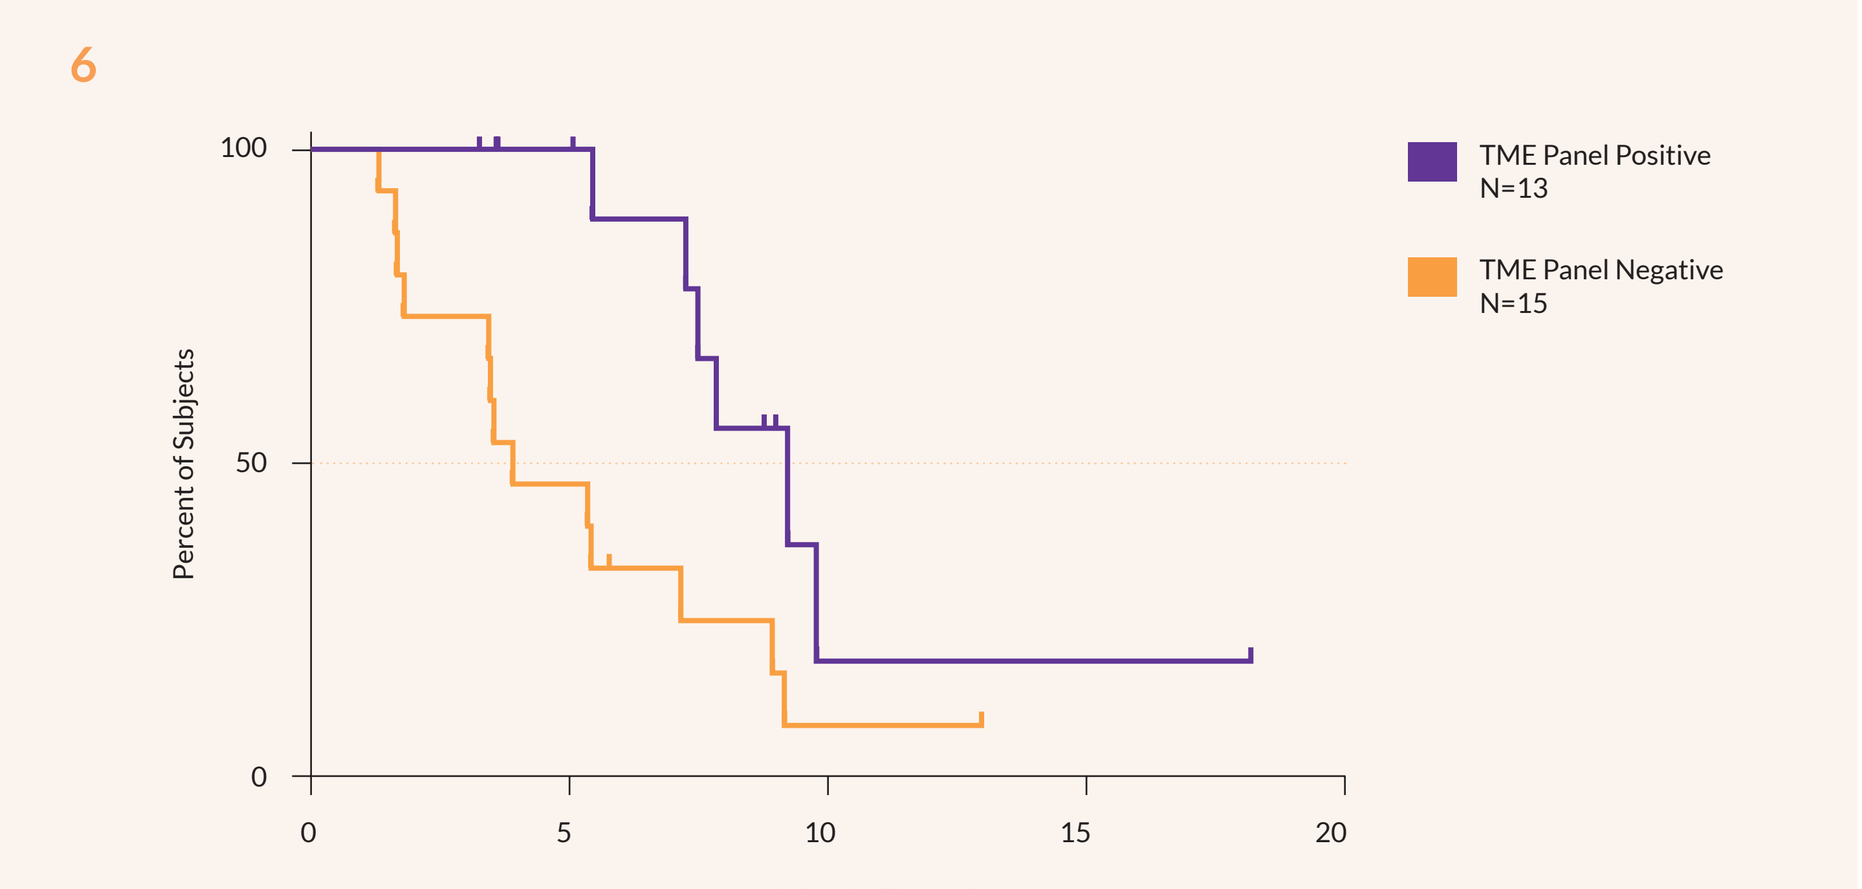
6
100
50
0
Percent of Subjects
0
5
10
15
20
Months Elapsed
TME Panel Positive N=13
TME Panel Negative N=15

Figure 6. Survival Benefit Conferred to Biomarker Positive Ovarian Patients
Kaplan Meier analysis of Progression Free Survival (PFS) for biomarker positive versus biomarker negative ovarian cancer subjects is shown for the navicixizumab phase 1b trial, retrospectively classified by the TME Panel-1 diagnostic. A statistically significant benefit in PFS of 9.2 versus 3.9 months was found (HR = 0.31, [0.125 to 0.784]).

6 Conclusions

- OncXerna's TME Panel-1 offers a unique approach to precision medicine based on a deep understanding of a patient's tumor microenvironment and dominant biology at the RNA level. This allows the potential for prospective identification of a larger group of responding patients and to pair those patients with clinical-stage therapies with known mechanism of action that directly address these biologies.
- The TME Panel is implemented as an artificial neural net algorithm that abstracts the dominant biologies of the tumor microenvironment from gene expression data, and classifies patients based on the TME phenotype of their cancer. The panel performs as well or better than gold standard industry biomarkers, demonstrating a relationship between clinical response and biomarker stratified patient populations.
- The TME Panel has retrospectively characterized tumor response to CPIs, immune activators and anti-angiogenic drugs. Bavituximab, in combination with pembrolizumab, achieves thus far durable responses creating an opportunity for patient subgroups that fail to respond to CPI monotherapy, and with navicixizumab, a bi-specific anti-angiogenic, the panel predicted an enhanced response in late-stage ovarian cancer patients.

References

- Cancer Genome Atlas Research Network. Comprehensive molecular characterization of gastric adenocarcinoma. Nature. 2014 Sep 11;513(7517):202-9.
- Cristescu R. Molecular analysis of gastric cancer identifies subtypes associated with distinct clinical outcomes. Nat Med. 2015 May;21(5):449-56.
- Lei Z. Identification of molecular subtypes of gastric cancer with different responses to PI3-kinase inhibitors and 5-fluorouracil. Gastroenterology. 2013 Sep;145(3):554-65.

Acknowledgments

The authors would like to thank Susan MacIntyre and Valerie Santos for their tireless work coordinating the exchange and QC of clinical data; Roman Lustrik for his help processing and curating gene expression and clinical data; and Eva Zupan Horacek for bringing the results to life through data visualization and poster design. Lastly, we are grateful to the Merck Bavituximab Collaboration Team for their ongoing contributions to the ONCG100 study, and helpful feedback on this presentation.

Copyright OncXerna Therapeutics, Inc., 2020