The Xerna™ TME Panel potentially predicts response to a combination of the TLR9 agonist vidutolimod and PD-1 inhibitor pembrolizumab in metastatic melanomas with prior anti-PD-1 treatment

OncXerna THERAPEUTICS™ | Genialis | REGENERON®

Mark Uhlik[1], Daniel Pointing[2], Luka Ausec[2], Miha Stajdohar[2], Robert Cvitkovic[2], Matjaz Zganec[2], Seema Iyer[1], Hong Liu[3], Art Krieg[3], Rafael Rosengarten[2], Laura Benjamin[1]

[1] OncXerna Therapeutics, Inc., Waltham, MA | [2] Genialis, Inc., Boston, MA | [3] Checkmate Pharmaceuticals, Inc., Cambridge, MA (acquired by Regeneron Pharmaceuticals, Inc., Tarrytown, NY on May 31, 2022)

AACR 2023 | Abstract Number **3270**

Correspondence: Mark Uhlik (muhlik@oncxerna.com)

ABSTRACT

Background:
Few therapeutic options exist for anti-PD-1 refractory, metastatic melanoma patients, and today's biomarkers are insufficient to aid in defining who should receive potential combinatorial immunotherapies. Results from a phase Ib, multicenter study (NCT02680184) showed that a combination of vidutolimod and pembrolizumab provided a best overall response of 23.5% in patients with metastatic or unresectable cutaneous melanoma who had received prior anti-PD-1 therapy. The Xerna™ TME Panel consists of an artificial neural net that utilizes the expression of ~100 genes involved in angiogenesis and tumor immune biologies to classify patient samples into one of four tumor microenvironment (TME) biomarker subtypes: Angiogenesis (A), Immune Active (IA), Immune Desert (ID), or Immune Suppressed (IS). The algorithm was trained and validated on gastric cancer data, it was predicted that the TME panel would be applicable in additional indications such as melanoma. We hypothesised that the IS subtype is predictive of vidutolimod + pembrolizumab benefit in this cohort compared to the other TME subtypes (A, IA, and ID).

Methods:
Total RNASeq was performed on formalin-fixed paraffin-embedded (FFPE) biopsies collected from a subset of patients prior to therapy (N=38) and 3 weeks post-initiation of therapy (N=10). Gene expression data was analyzed using the Xerna TME Panel algorithm to assign a TME subtype. Correlational analyses between TME subtypes, response to therapy, and other hallmark gene signatures were performed.

Results:
Overall response rate in the pretreament cohort available for biomarker analysis was 26%, comparable to the entire vidutolimod/pembrolizumab arm. The cohort had a skewed distribution of TME subtypes with high prevalence of IS (34%) and ID (45%), indicative of immune therapy-refractory biologies. An overall response of 54% was observed in the IS subtype, compared with 12% in the other subtypes combined. Comparison with other "hallmark" gene signatures confirmed enrichment of immune and angiogenesis biologies in the IS subtype, but none of these individual hallmark signatures were found to differentiate between responders and non-responders. The Xerna TME Panel demonstrated superior classification performance across all criteria compared to a baseline classifier, including accuracy (0.76 vs. 0.62), sensitivity (0.70 vs. 0.27) and specificity (0.79 vs. 0.74). Among 10 matched post-treatment samples, 70% revealed a change in TME subtype compared to their pre-treatment status. Three of the post-treatment samples represented changes from an immune-low subtype to an immune-high subtype, including one complete responder with a pre-treatment ID that changed to IA while on therapy, illustrating how the TME Panel may be used to interpret the mechanism of drug response.

Conclusions:
The Xerna TME Panel shows potential activity as a predictive and pharmacodynamic biomarker for the combination of vidutolimod and pembrolizumab in anti-PD-1 refractory melanoma patients.

BACKGROUND

Overview of Xerna TME Panel

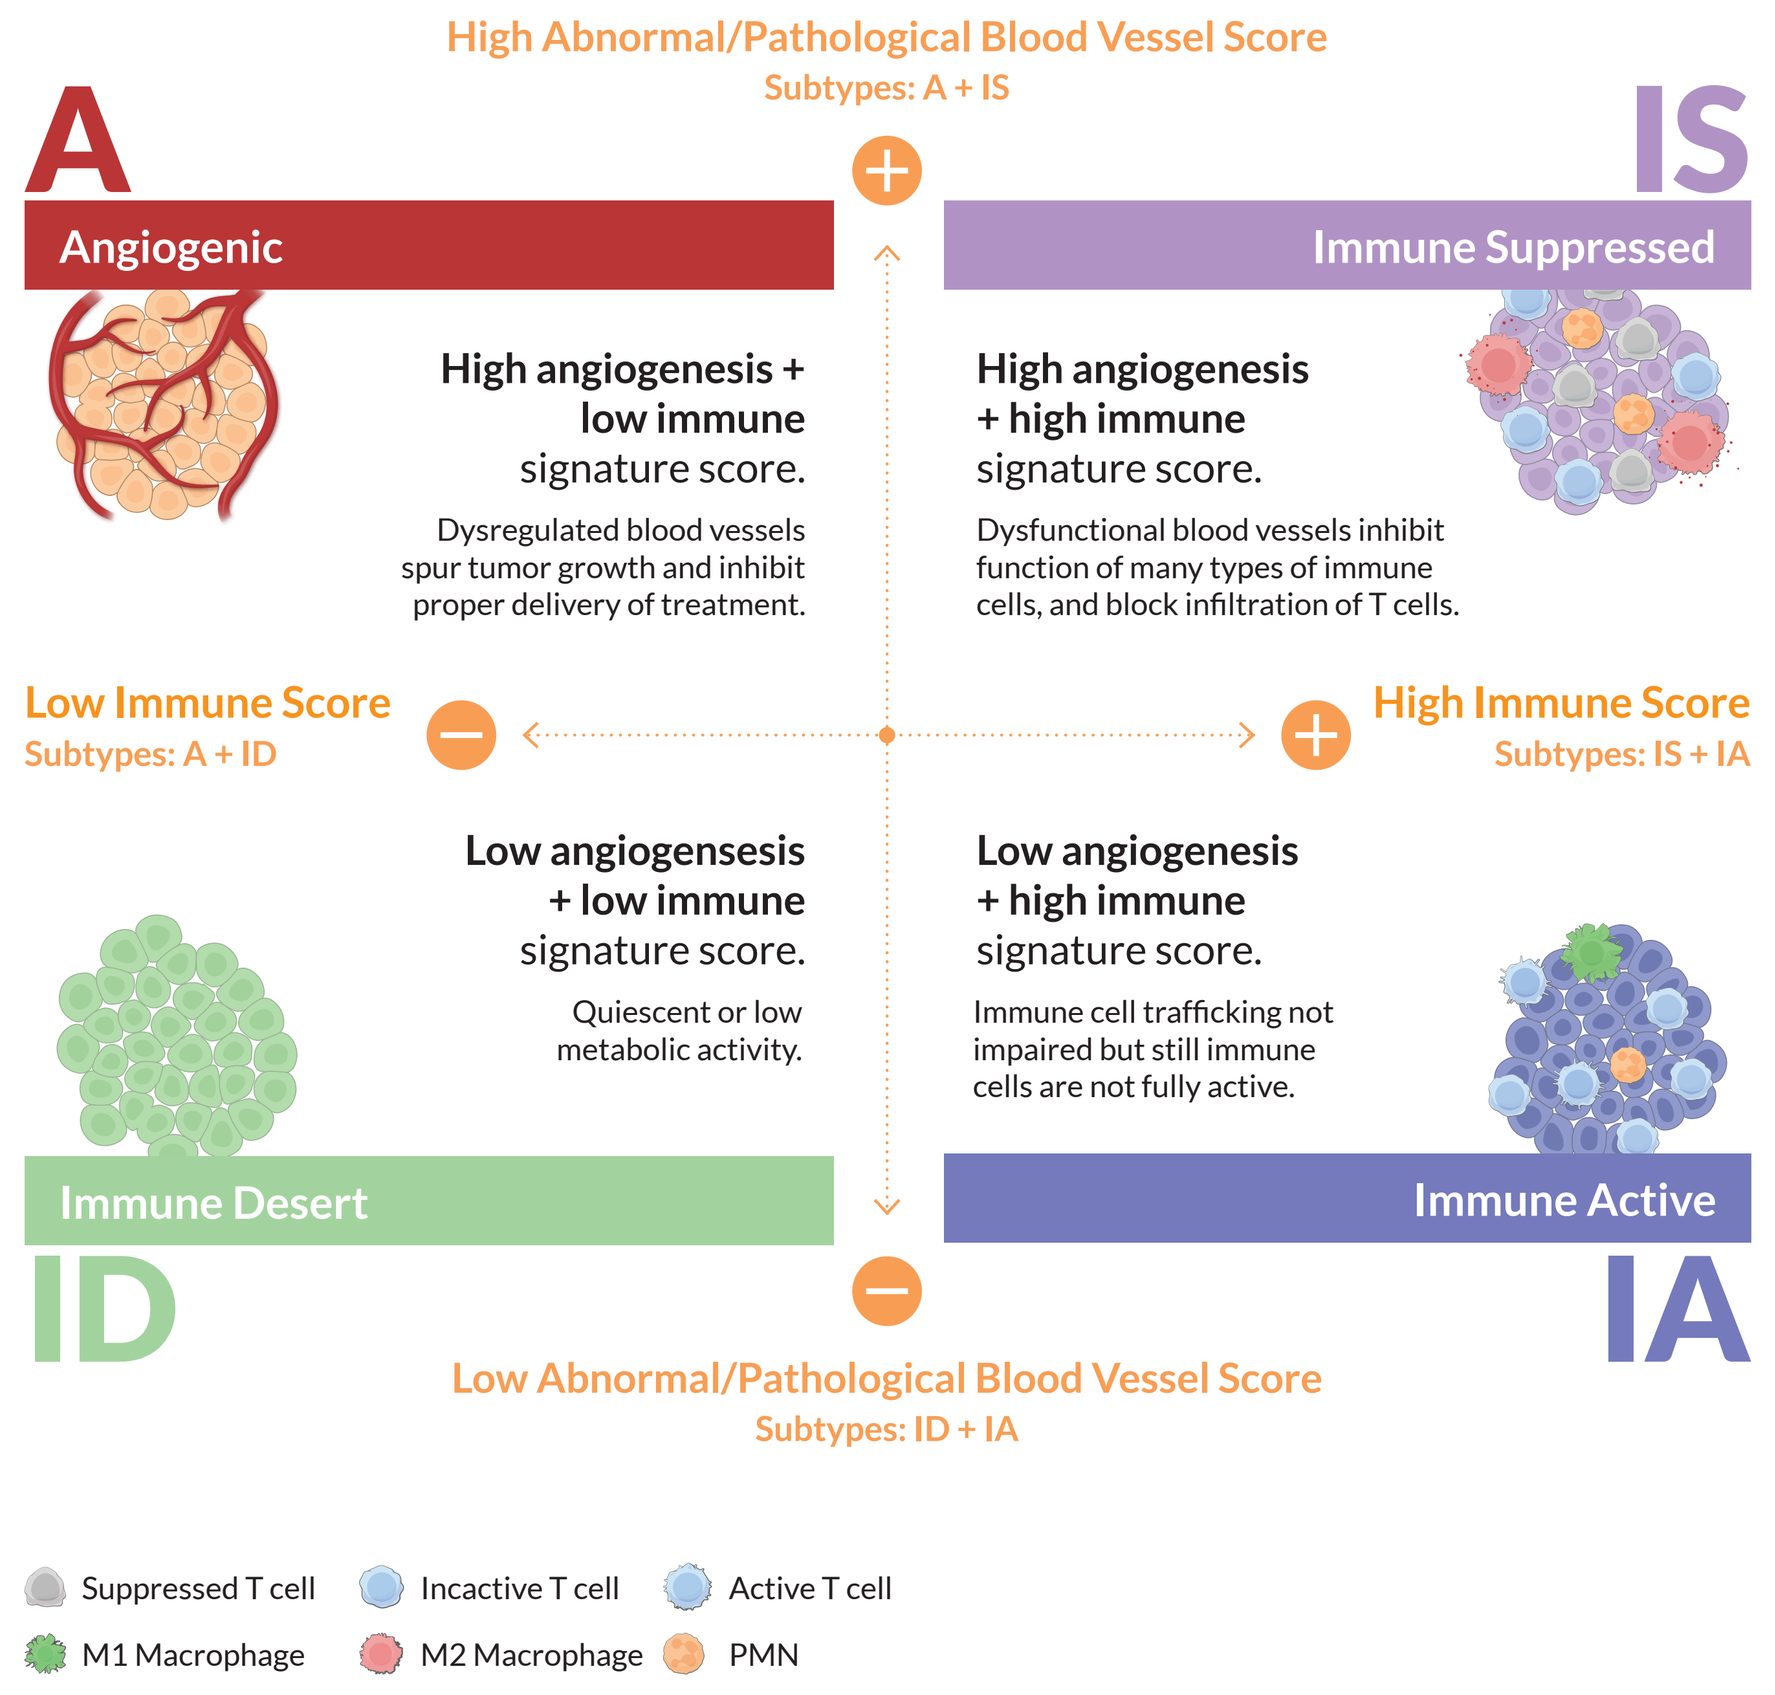
High Abnormal/Pathological Blood Vessel Score
Subtypes: A + IS
A
Angiogenic
IS
Immune Suppressed
High angiogenesis + low immune signature score.
Dysregulated blood vessels spur tumor growth and inhibit proper delivery of treatment.
High angiogenesis + high immune signature score.
Dysfunctional blood vessels inhibit function of many types of immune cells, and block infiltration of T cells.
Low Immune Score
Subtypes: A + ID
High Immune Score
Subtypes: IS + IA
Low angiogensesis + low immune signature score.
Quiescent or low metabolic activity.
Low angiogenesis + high immune signature score.
Immune cell trafficking not impaired but still immune cells are not fully active.
Immune Desert
Immune Active
ID
IA
Low Abnormal/Pathological Blood Vessel Score
Subtypes: ID + IA
Suppressed T cell
Incactive T cell
Active T cell
M1 Macrophage
M2 Macrophage
PMN

The Xerna TME Panel is a novel diagnostic assay that uses formalin-fixed paraffin-embedded (FFPE) tissue-derived RNA gene expression data based on ~100 genes to classify patients into dominant biologies of the tumor microenvironment (TME). The input gene signature represents angiogenic and immunogenic properties of stromal biology, and the machine learning neural network that comprises the Xerna TME Panel algorithm has learned interactions between these critical processes.

The Xerna TME Panel can be used to classify a patient's tumor microenvironment along an immune and angiogenic axis, resulting in one of four TME phenotypes—***Angiogenic* (A)**, ***Immune Active* (IA)**, ***Immune Desert* (ID)**, and ***Immune Suppressed* (IS)** (figure above). Each class, or combinations of classes, can be predictive of treatment outcome with various targeted therapies, including anti-angiogenics and checkpoint inhibitors, as well as novel drugs targeting the intersection of these biologies.

Vidutolimod Mechanism-of-Action

Vidutolimod can help to restore the antitumor T-cell response:

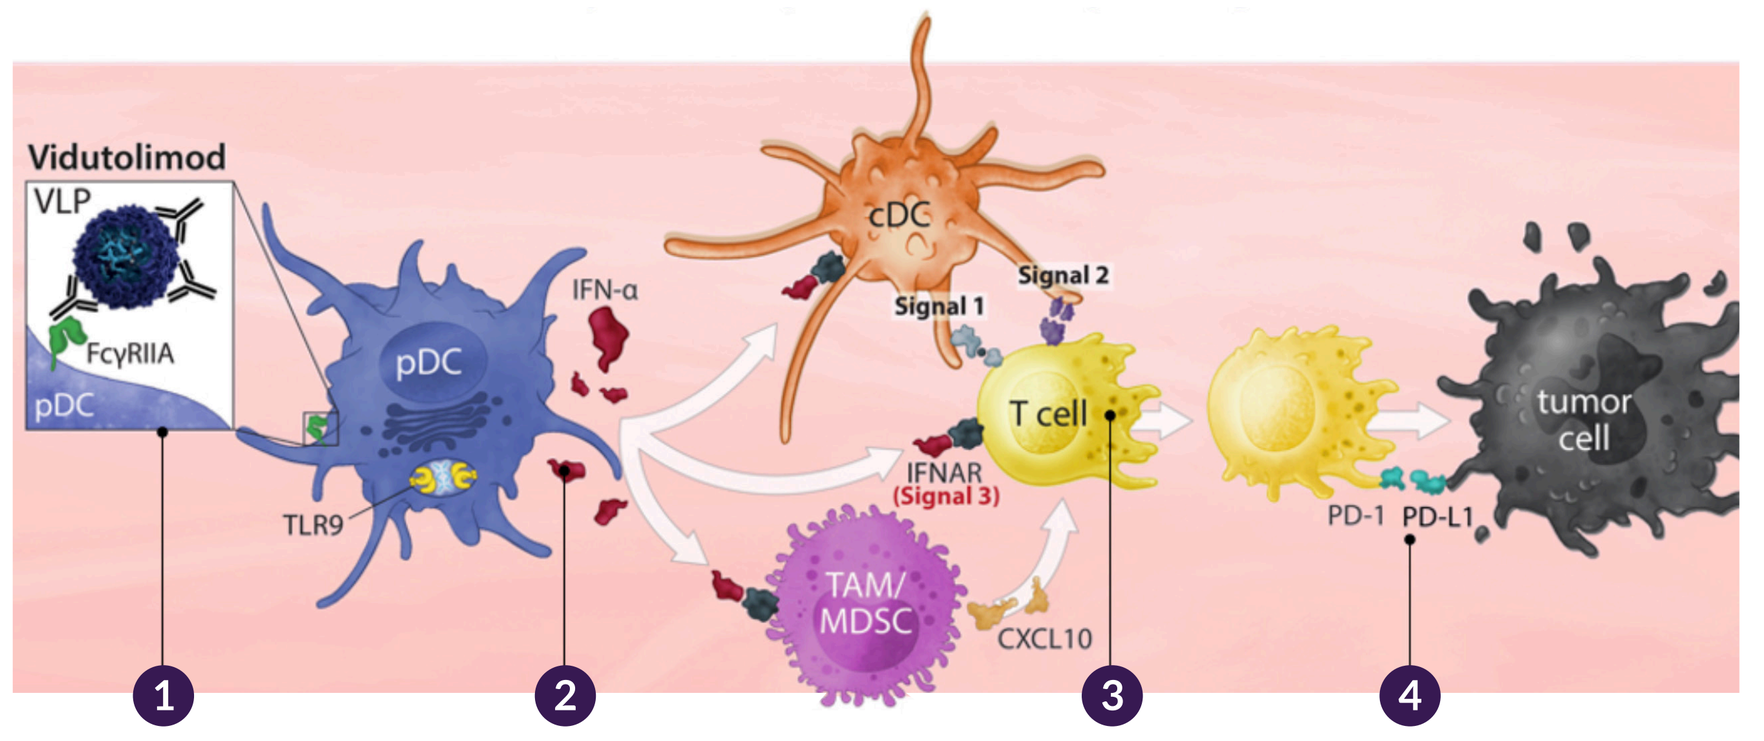
Vidutolimod
VLP
FcγRIIA
pDC
pDC
TLR9
IFN-α
cDC
Signal 1
Signal 2
T cell
IFNAR
(Signal 3)
TAM/
MDSC
CXCL10
PD-1
PD-L1
tumor
cell
1
2
3
4

1. Antibody-bound vidutolimod is taken up by pDC and activates TLR9
2. Activated pDC recruit and stimulate other immune cells; Type I IFN enhances T cell activation directly and indirectly, e.g., CXCL10 primary PD marker for clinical response
3. Neoantigen-specific T cells mediate systemic tumor regression
4. Blockade of PD-1 improves duration of T cell response; rationale for combo therapy

APPROACH

GSVA Gene Signatures are Not Able to Differentiate Responders and Non-Responders

GSVA enrichment scores were plotted for each patient sample and grouped according to their best overall response to drug. Data is shown for the most relevant hallmark signatures to each cohort (i.e. angiogenesis-related signatures for cohorts treated with anti-angiogenic agents). Box-whisker plots show each sample as an open circle, with the "x" representing the mean, and the horizontal line representing the median of each group.

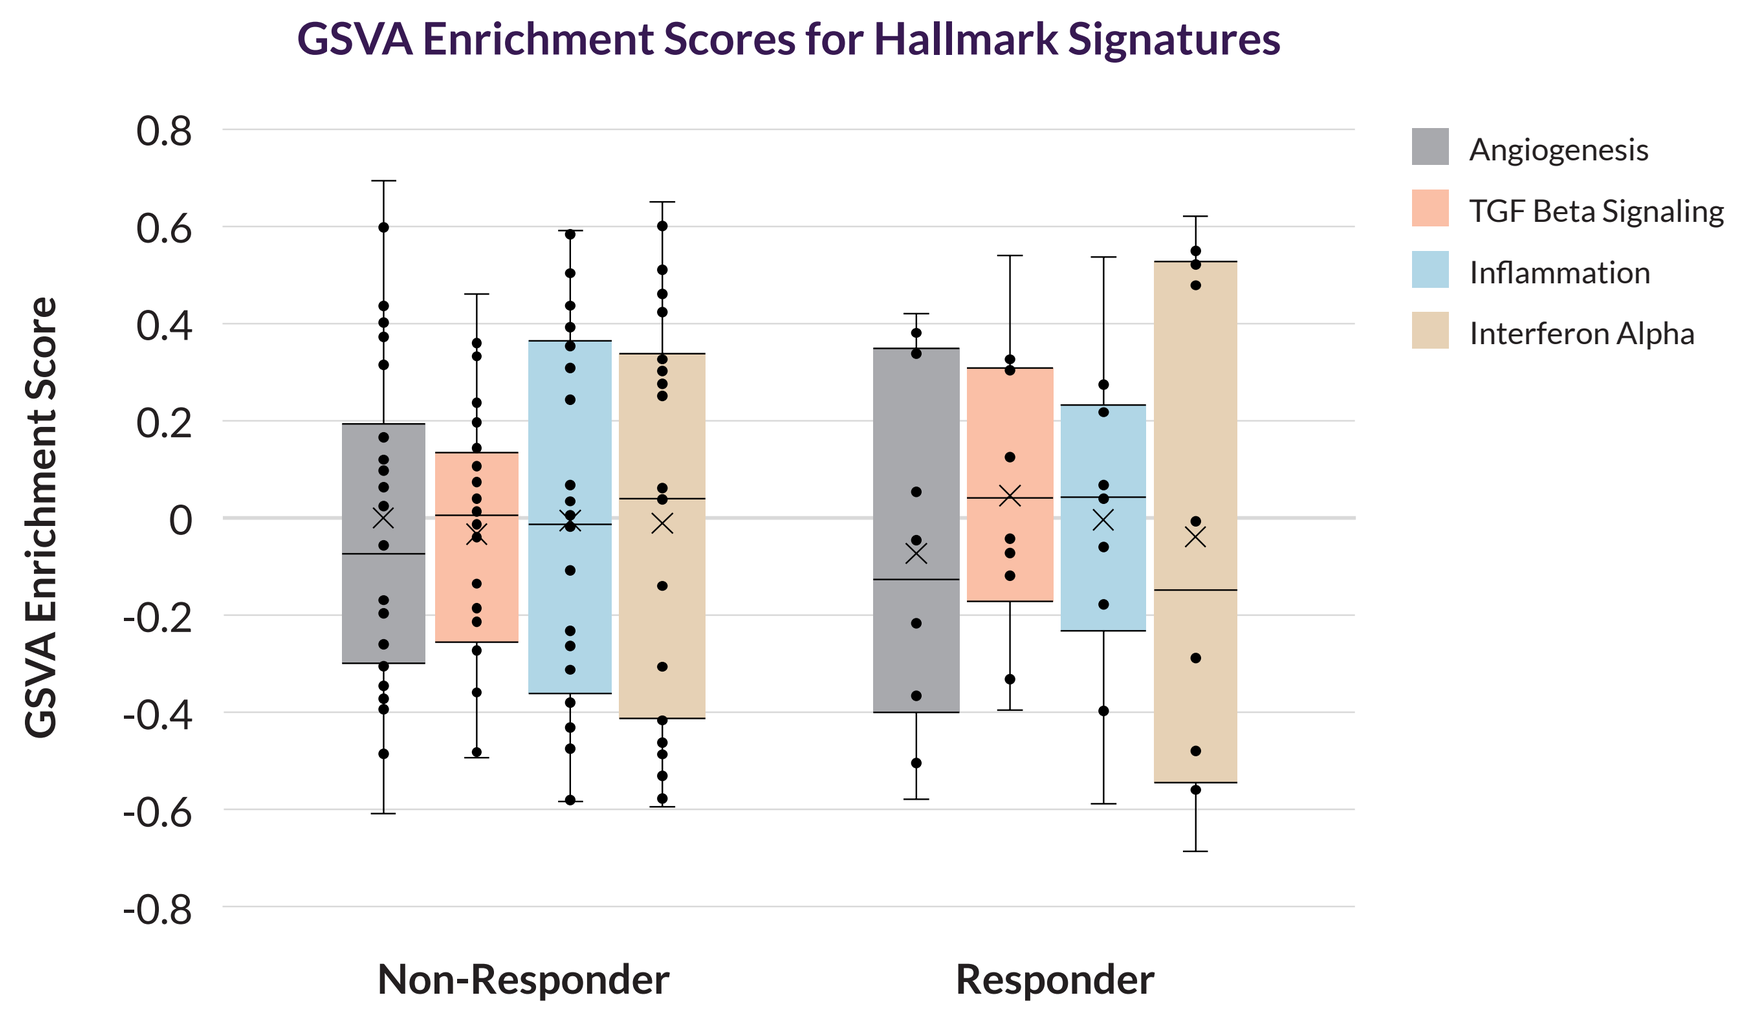
GSVA Enrichment Scores for Hallmark Signatures
GSVA Enrichment Score
0.8
0.6
0.4
0.2
0
-0.2
-0.4
-0.6
-0.8
Non-Responder
Responder
Angiogenesis
TGF Beta Signaling
Inflammation
Interferon Alpha

Xerna TME Panel Workflow

Gene expression data (SAMPLE) is analyzed through a machine learning-based artificial neural net algorithm (MODEL) which provides a single subtype designation (OUTPUT) that is based on probabilized scores of the TME subtypes (CONFIDENCE). Shown here is a representation of a single tissue sample analyzed by the model with its subtype designation "ID" and visualization on a latent space plot highlighted as a green circle.

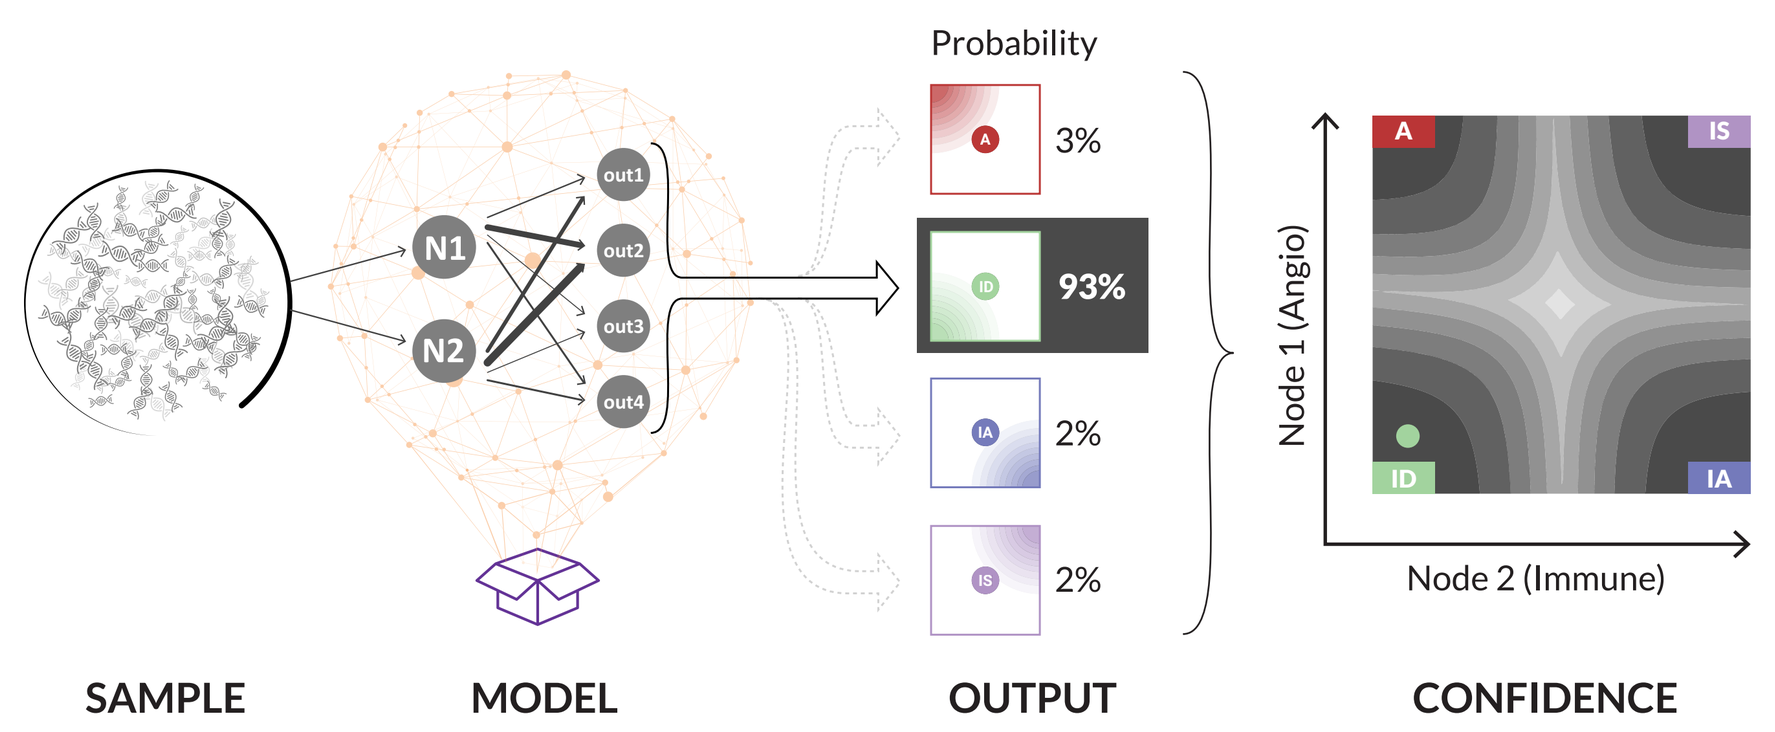
N1
N2
out1
out2
out3
out4
Probability
3%
93%
2%
2%
Node 1 (Angio)
Node 2 (Immune)
A
IS
ID
IA
SAMPLE
MODEL
OUTPUT
CONFIDENCE

STUDY DETAILS

- Ph1b study (NCT02680184) of advanced melanoma with prior progression on one or more immune checkpoint therapies (N=38 for pre-treatment biomarker analysis, N=10 for on-treatment biomarker analysis)
- Treatment with TLR9 agonist Vidutolimod + anti-PD-1 Pembrolizumab
- ORR data was available for assessment of Xerna TME panel biomarker predictive potential
- Tumor samples were biopsies collected just prior to initiating trial therapy (pre-treatment samples) or at 3 weeks after 1st dose (on-treatment samples)
 - WTS was performed on FFPE
 - Analyzed in the Xerna TME panel
- **TME Immune Suppressed subtype (IS) was hypothesized to derive the most clinical benefit.**

RESULTS

Patient Cohort is Highly Enriched for IS and ID Xerna TME Subtypes

Xerna TME analysis of this cohort revealed an enrichment of immune suppressed (IS) and immune desert (ID) subtypes. This is consistent with a hypothesis that previous exposure to and progression on immune checkpoint inhibitors (ICI) correlates with tumor microenvironments that are populated with exhausted or suppressive immune cells or are largely devoid of immune infiltration.

Distribution of Xerna TME Subtypes

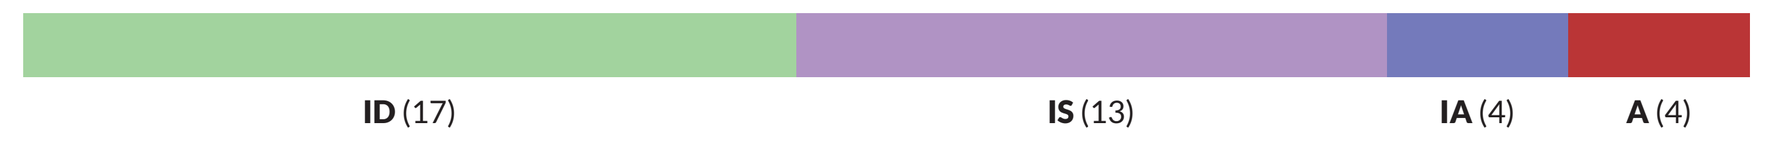
ID (17)
IS (13)
IA (4)
A (4)

Gene Set Variation Analysis (GSVA) showing enrichment of various pathways (rows) for each patient in the cohort (columns). Patients are grouped on the x-axis by TME subtype and tumor response based on RECIST criteria, shown according to the legend. GSVA signatures are grouped on the y-axis by general biological class with angiogenesis-related biology on top, immune-related biology in middle, and cell cycle and proliferation-related biology at the bottom. The IS TME subtype that correspond to the biomarker-positive status for this cohort is boxed in gray color.

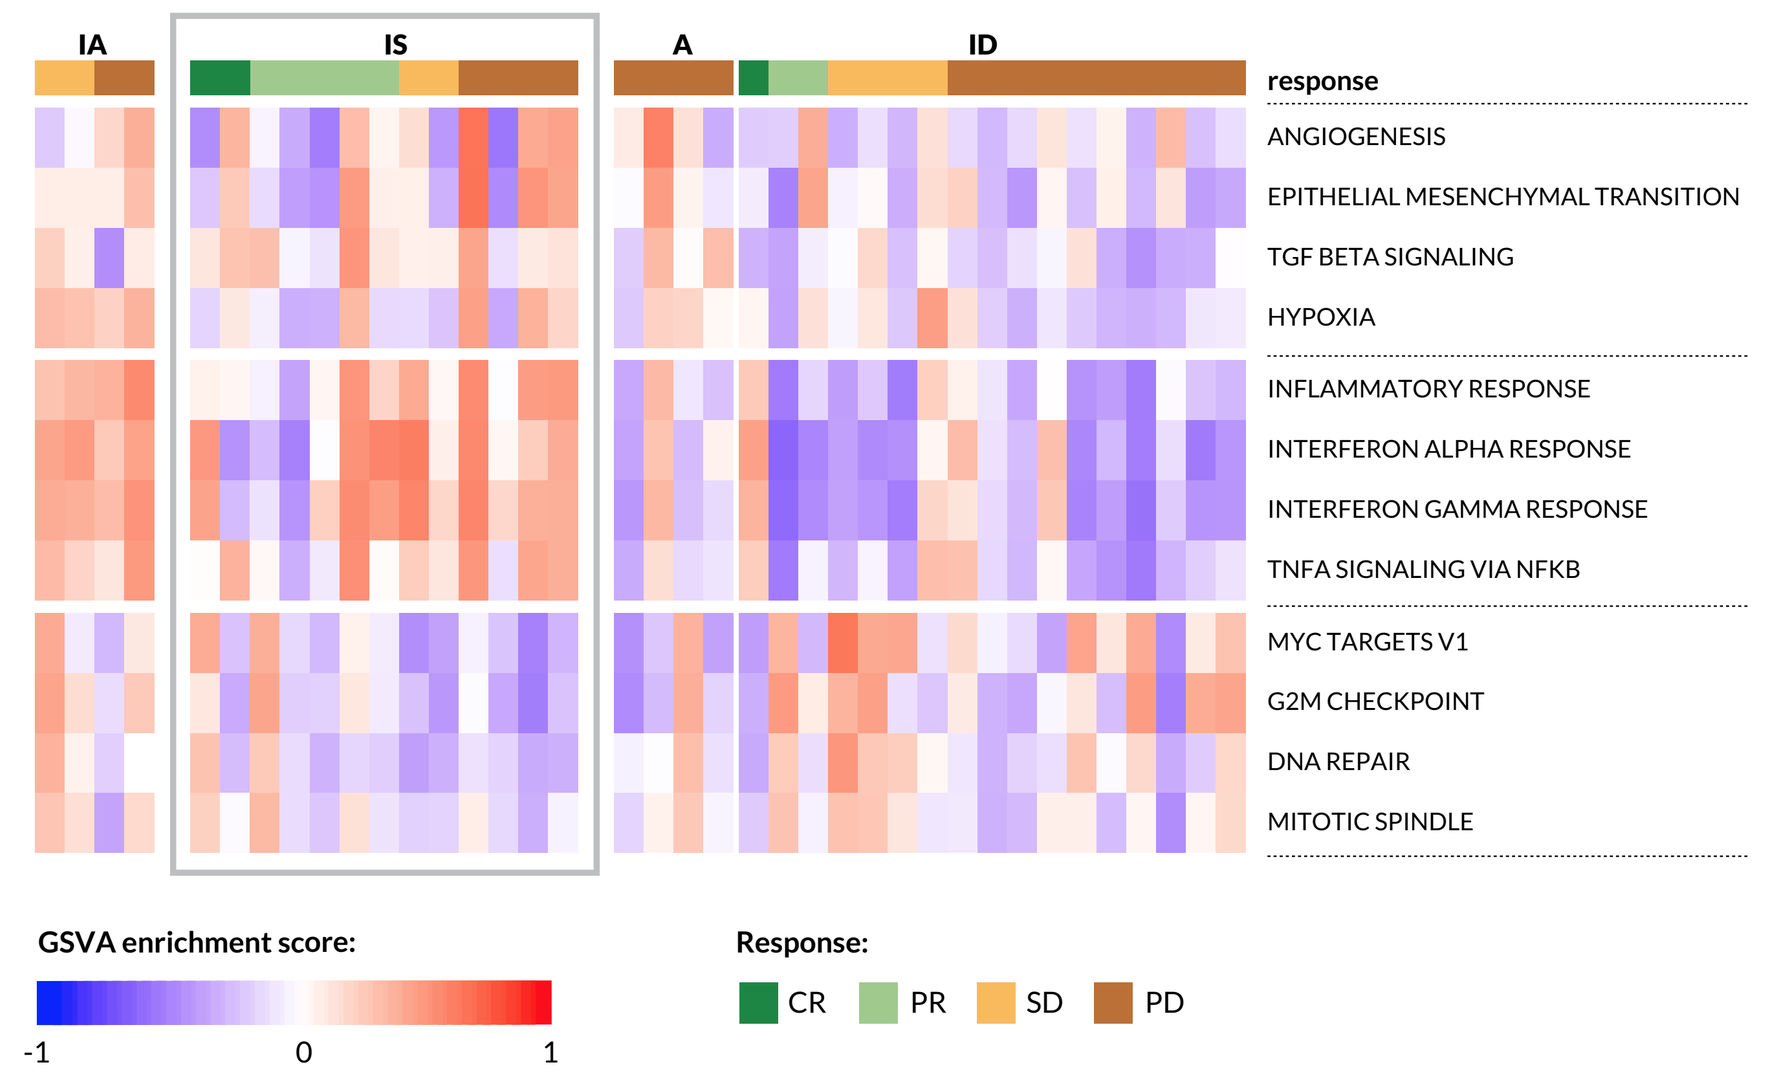
IA
IS
A
ID
response
ANGIOGENESIS
EPITHELIAL MESENCHYMAL TRANSITION
TGF BETA SIGNALING
HYPOXIA
INFLAMMATORY RESPONSE
INTERFERON ALPHA RESPONSE
INTERFERON GAMMA RESPONSE
TNFA SIGNALING VIA NFKB
MYC TARGETS V1
G2M CHECKPOINT
DNA REPAIR
MITOTIC SPINDLE
GSVA enrichment score:
-1
0
1
Response:
CR
PR
SD
PD

IS Subtype is Enriched with Responders, Particularly in Those with 1 Prior ICI

Stacked bar charts show best overall responses sorted by Xerna TME subtype and prior ICIs received.

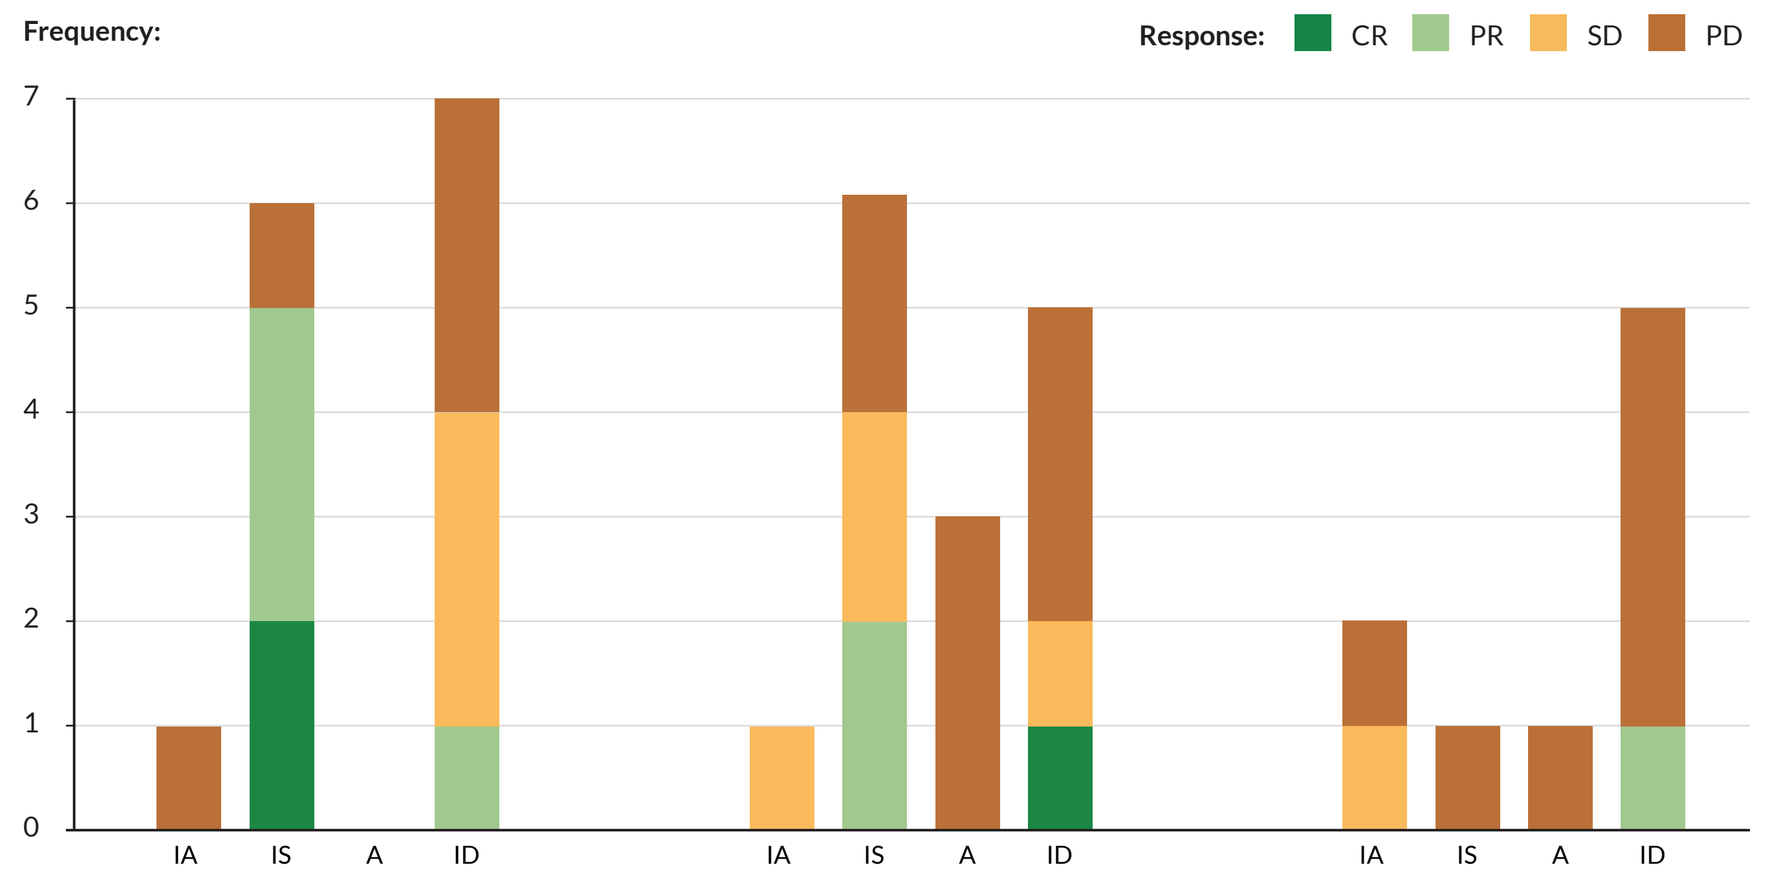
Frequency:
Response:
CR
PR
SD
PD
IA
IS
A
ID

1 prior ICI (N = 14)	2-3 prior ICIs (N = 15)	>3 prior ICIs (N = 9)
6/14 total responders 42.9% ORR	3/15 total responders 20% ORR	Reduced prevalence of IS subtype
5/6 IS had CR/PR 83.3% ORR	2/6 IS had CR/PR 33.3% ORR	1/9 responders (ID patient) 11.1% ORR

Xerna TME Panel Demonstrates Predictive Potential for Vidutolimod + Pembrolizumab

(A) Patient tumor samples from the vidutolimod + pembrolizumab cohort projected on the latent space of the TME Panel classifier. The latent space is a two-dimensional representation of the two neurons in the hidden layer of the model, with neuron 1 consisting of genes linked with immunogenic biology as the x-axis (Immune Score) and neuron 2 similarly with genes synonymous with angiogenesis as the y-axis (Angio Score). The gray contours create a probability gradient, as indicated in the legend. Each glyph is a patient sample, colored according to RECIST score, according to the legend. **(B)** Tumor response for each Xerna TME subtype tabulated based on RECIST score.

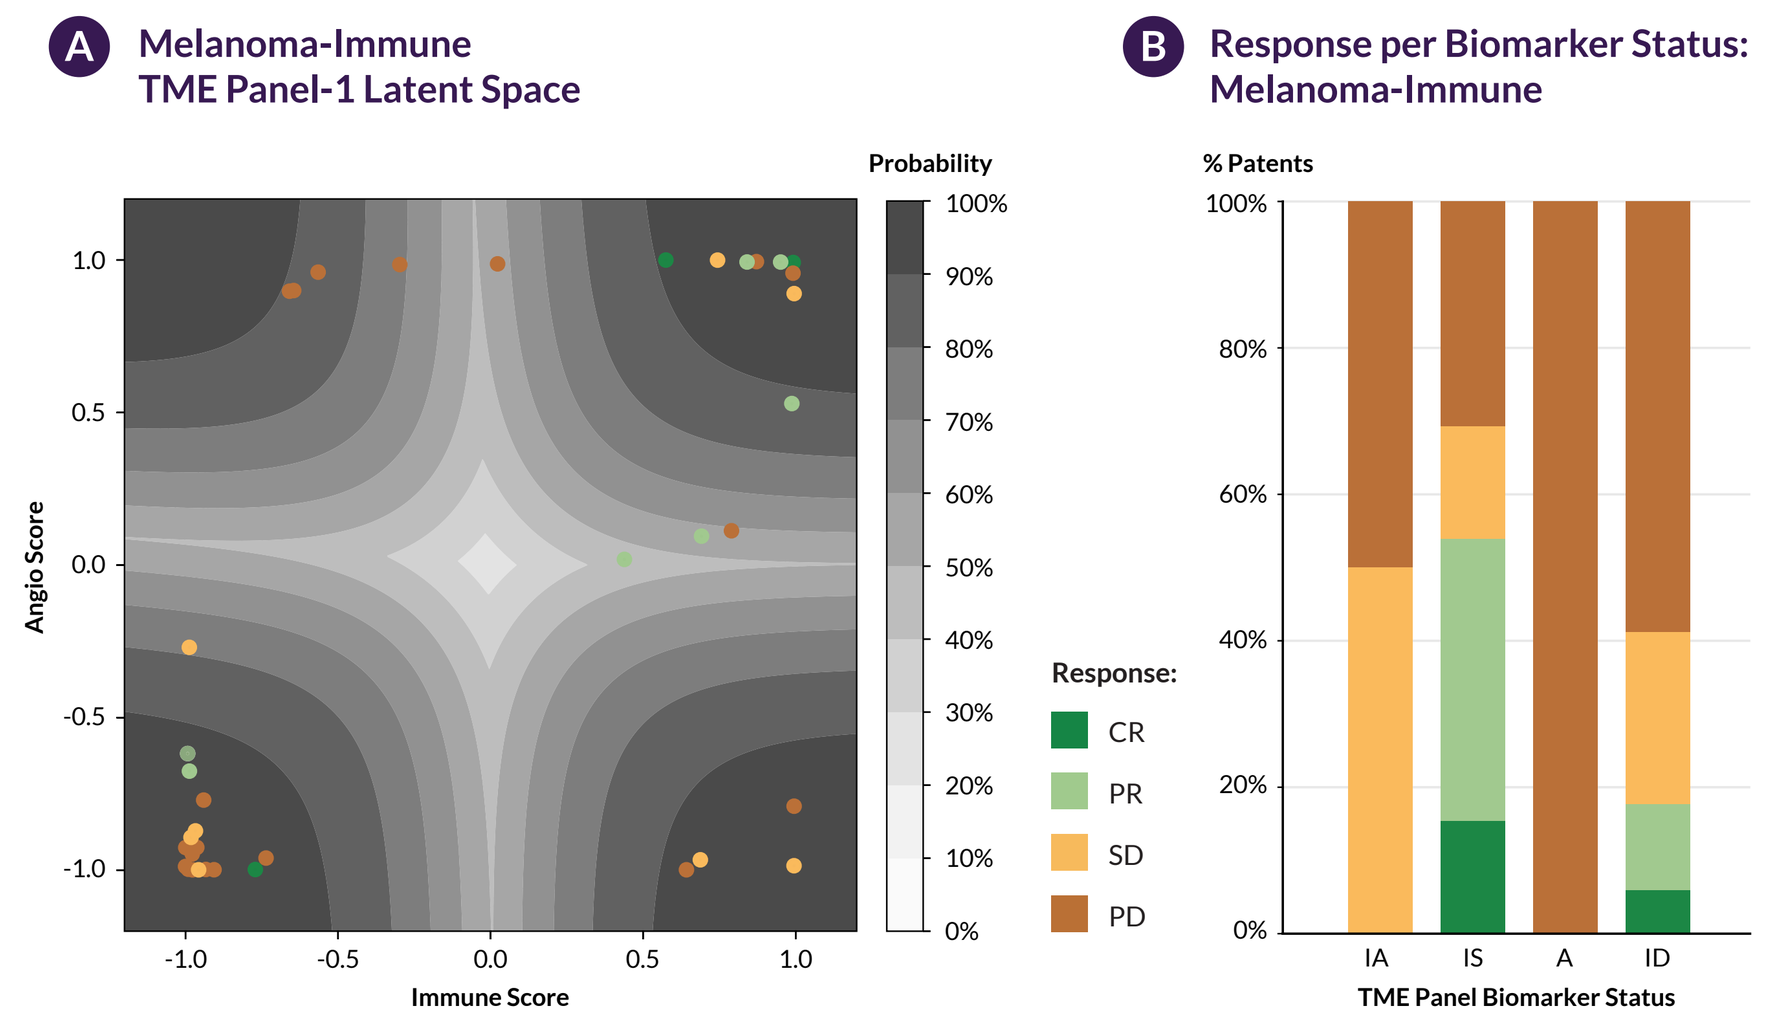
A
Melanoma-Immune
TME Panel-1 Latent Space
Probability
Angio Score
Immune Score
Response:
CR
PR
SD
PD
B
Response per Biomarker Status:
Melanoma-Immune
% Patents
TME Panel Biomarker Status
IA
IS
A
ID

Key Takeaways:

- High prevalence of Immune Desert (ID; 45%) and Immune Suppressed (IS; 34%) subtypes in cohort
- ORR of 26% (10/38) in entire cohort
- ORR of 54% (7/13) in IS subtype (biomarker positive)
- ORR of 12% (3/25) in non-IS subtypes (biomarker negative)
- **Enrichment of ~4.5X between biomarker-positive vs. –negative**

Xerna TME Panel Performance Characteristics for Melanoma Cohort

Vidu + Pembro (N=38) | B+ = TME IS | ORR(B+) = 53.8% | ORR(B-) = 12.0% | ORR enrichment = 4.5x

Biomarker/ Model	ACC	AUC ROC	F1	Sensi-tivity	Speci-ficity	Precision/ PPV	NPV
TME	0.76 (29/38)	0.75	0.61	0.7 (7/10)	0.79 (22/28)	0.54 (7/13)	0.88 (22/25)
Baseline	0.62	0.50	0.27	0.27	0.74	0.27	0.74

A simple baseline classifier served to represent the null model. The baseline classifier randomly samples the class based on prior class probabilities and simulates drug response without a biomarker. Column headers represent standard machine learning performance metrics:

- ***Accuracy (ACC):*** *Number of correct predictions / Total number of predictions*
- ***Area Under the Receiver Operator Curve (AUROC):*** *Degree to which model is capable of distinguishing between classes*
- ***Sensitivity (Recall):*** *True biomarker responders / Total actual responders*
- ***Specificity:*** *True biomarker non-responders / Total actual non-responders*
- ***Positive Predictive Value (PPV or Precision)****: True biomarker responders / Total predicted biomarker responders*
- ***Negative Predictive Value (NPV):*** *True biomarker non-responders/ Total predicted biomarker non-responders*

The Xerna TME Panel Reveals Pharmacodynamic Effects of Treatment

Analysis of matched pre and post treatment samples. Post treatment samples were taken on Day 1 of Week 3 of treatment. The arrows demonstrate how the sample has changed after treatment. Blue circles are pre-treatment and orange circles are matched on-treatment.

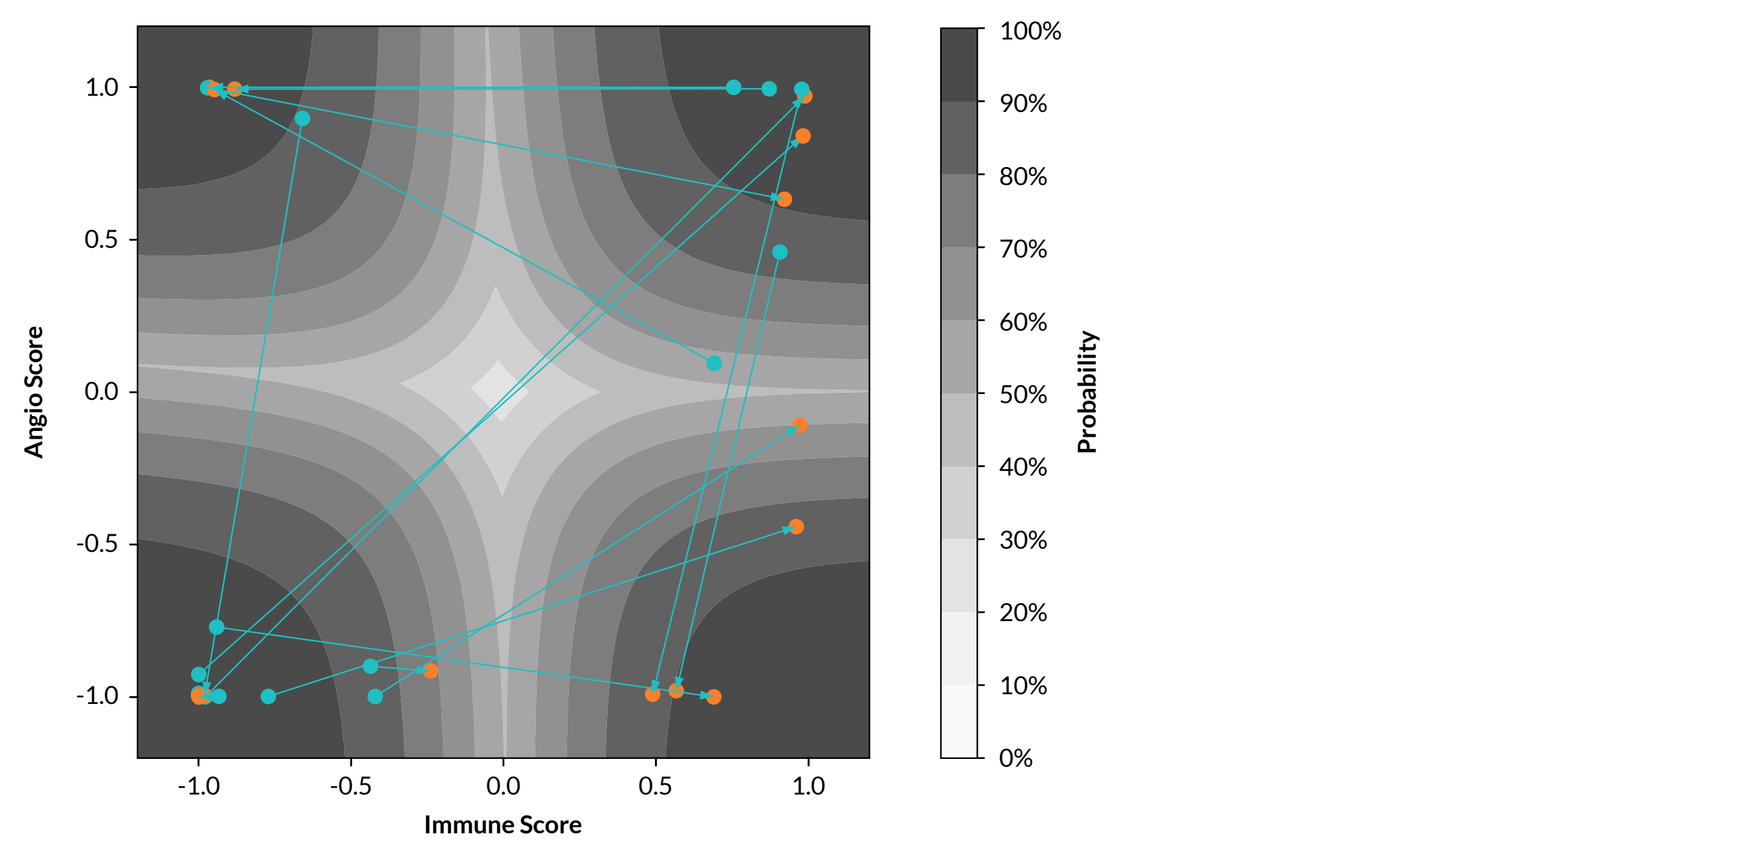
Angio Score
Immune Score
Probability

	Pre-Tx TME Subtype	Post-Tx TME Subtype	Response
Subject 1	IS	IA	PR
Subject 2	ID	IA	CR
Subject 3	ID	IS	SD
Subject 4	ID	IS	PD
Subject 5	ID	IA	PD
Subject 6	ID	ID	PD
Subject 7	ID	ID	PD
Subject 8	A	ID	PD
Subject 9	IS	A	PR
Subject 10	IS	A	PD

SUMMARY AND CONCLUSIONS

The Xerna TME Panel is a machine-learning based, artificial neural net model using RNA-Seq technology for RNA derived from FFPE tumor tissue. The gene signature identifies the dominant biology of the tumor microenvironment and assigns samples to therapeutically actionable tumor subtypes defined by angiogenesis and immune gene expression.

High prevalence of IS and ID subtypes were observed in this post-anti-PD-1 therapy melanoma cohort.

Xerna TME Panel demonstrated predictive potential for combination immunotherapies – Vidutolimod + Pembrolizumab.

ORR enrichment of 4.5X between biomarker-positive (IS) and biomarker-negative (A, IA, ID) subtypes.

Superior predictive performance compared to baseline model; other GSVA hallmark signatures are not predictive.

Pharmacodynamic effects were observed in post-treatment samples - shift from non-immune to immune subtypes (including one pre-treatment ID complete responder).